AMCP 2025

Mar 31 – Apr 3
HOUSTON, TX

Misdiagnoses of Autoimmune Type 1 Diabetes (T1D) or Latent Autoimmune Diabetes in Adults (LADA) as Type 2 Diabetes (T2D) in Adults: A Systematic Literature Review (SLR)

Laura Wilson[1], Mariam Hanna[2], Gaurang Nazar[3], Divya Pushkarna[3], Boris Breznen[3]

[1]Sanofi, Bridgewater, NJ, USA; [2]Sanofi, Morristown, NJ, USA; [3]Evidinno Outcomes Research Inc., Vancouver, BC, Canada

To download e-poster scan the QR code

INTRODUCTION

- Autoimmune type 1 diabetes (T1D) has traditionally been considered a childhood or adolescent-onset condition. However, recent epidemiological data reveal that over 50% of new T1D cases occur in adults.[1]
- Additionally, up to 40% of adults aged 30 or older who are initially misdiagnosed with type 2 diabetes (T2D) may, in fact, have autoimmune T1D.[2,3]
- Such misdiagnoses represent critical missed opportunities for early intervention, potentially delaying disease progression and impacting treatment outcomes.

OBJECTIVE

To review the prevalence of misdiagnosis of T1D or latent autoimmune diabetes in adults (LADA) as T2D in adults and explore the role of diagnostic tests and healthcare professionals in cases of such misdiagnosis.

METHODS

- A systematic literature review (SLR) was conducted by searching Embase and MEDLINE® from database inception to April 3, 2024, following Cochrane Handbook guidelines.[4] Eligible studies were observational studies reporting misdiagnosis of autoimmune T1D or LADA as T2D in adults (≥18 years).
- Conference abstracts (2022–2024) from the American Diabetes Association (ADA) and European Association for the Study of Diabetes (EASD) were also reviewed.
- Two independent investigators screened abstracts, performed full-text review, and data extraction. Findings were summarized according to the Preferred Reporting Items for Systematic Reviews and Meta-Analyses (PRISMA) guidelines.[5]
- Study quality was assessed using the Joanna Briggs Institute (JBI) checklist.[6]

RESULTS

- A total of 5,870 abstracts were identified: 3,589 from Embase, 2,281 from MEDLINE®, and 4 from citation searching. After screening, 16 studies were included in the SLR (**Figure 1**).
- Most studies were from UK (6 studies)[7-12] and USA (2)[13-14] followed by one study each from Australia,[15] Tanzania,[16] India,[17] Spain,[18] Japan,[19] Germany,[20] France,[21] and Jamaica.[22]
- Study populations included T1D (5 studies), T2D (3), T1D and T2D (7), and LADA (1). The number of recruited patients ranged from 10 to 38,344 (median: 316), with patient age ranging from 43 to 67 years (median: 52.5).
- Nearly 45% of participants were female, 95.2% were Caucasian, and the median duration since T1D/LADA diagnosis was 10.4 years. T1D/LADA misdiagnoses were assessed using patient surveys/medical records, while diagnostic tests were used for T2D patients.

Misdiagnoses rates

- Misdiagnoses rates for T1D as T2D ranged from 3% - 47% (median: 22%, 9 studies) (**Table 1).** Among clinically diagnosed T1D, rates ranged from 3% to 47% (median: 23%, 6 studies).

RESULTS

Figure 1: PRISMA flow diagram

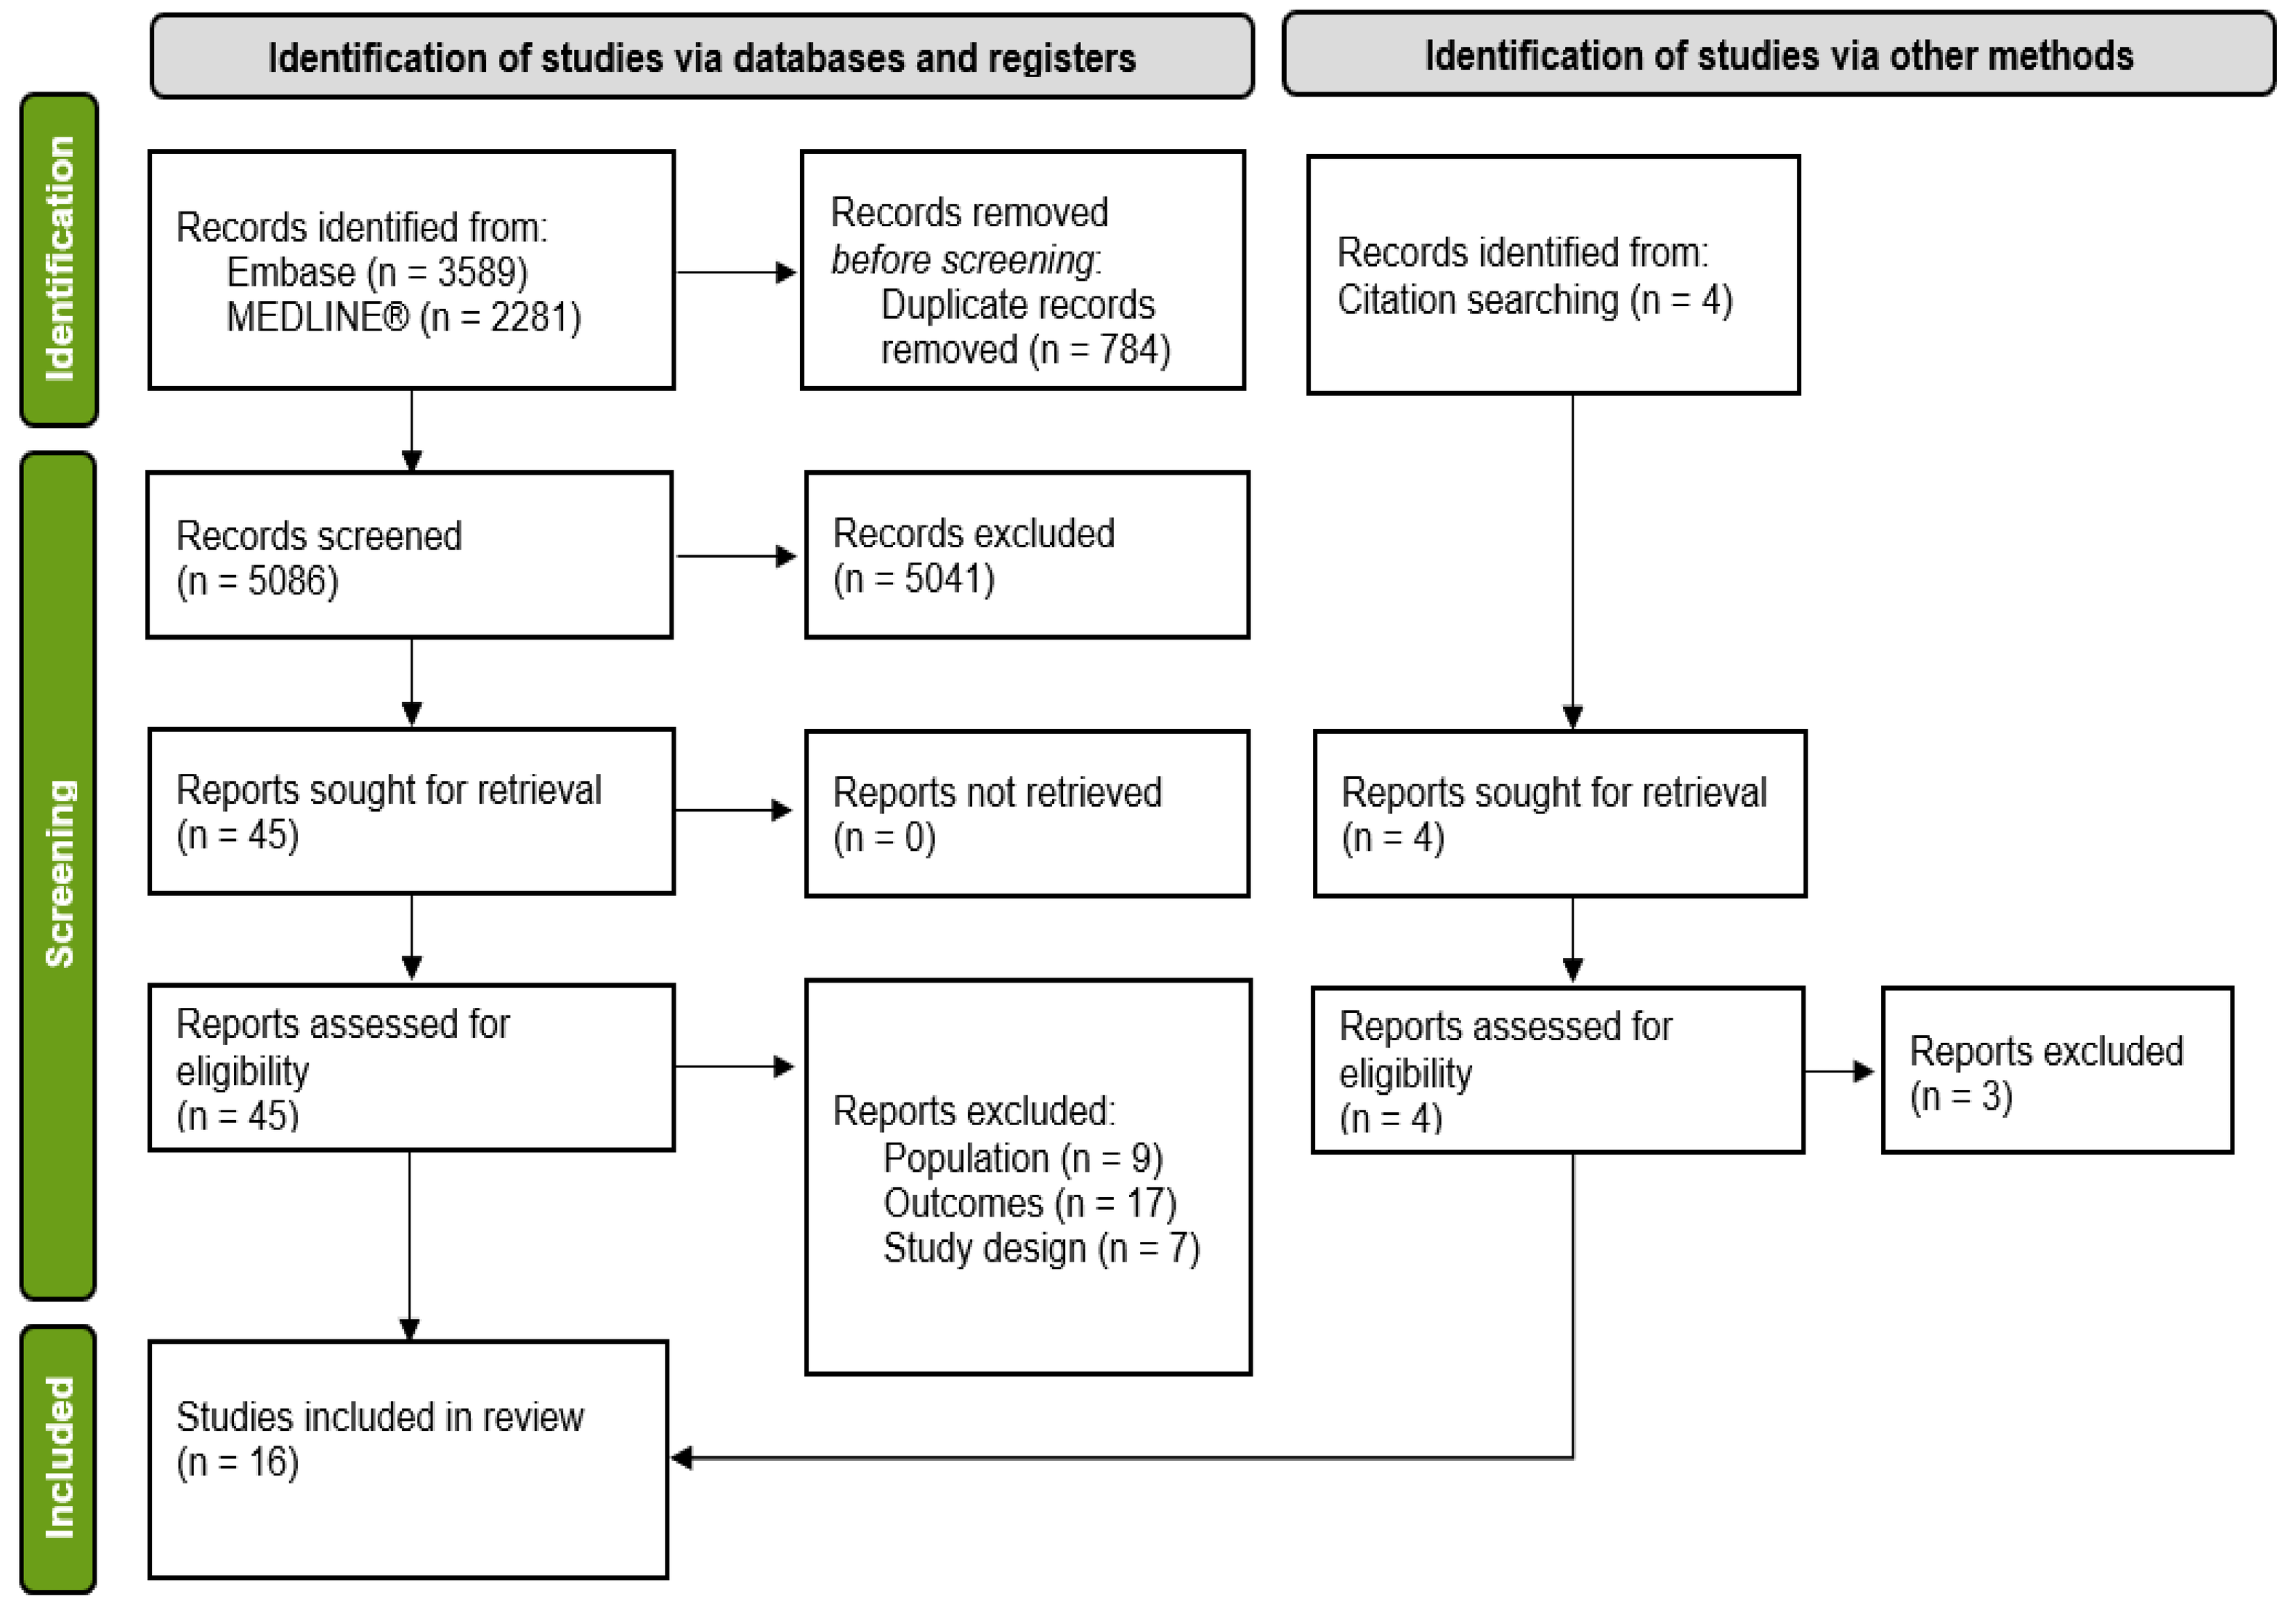

Identification of studies via databases and registers
Identification of studies via other methods
Identification
Screening
Included
Records identified from:
Embase (n = 3589)
MEDLINE® (n = 2281)
Records removed before screening:
Duplicate records removed (n = 784)
Records identified from:
Citation searching (n = 4)
Records screened
(n = 5086)
Records excluded
(n = 5041)
Reports sought for retrieval
(n = 45)
Reports not retrieved
(n = 0)
Reports sought for retrieval
(n = 4)
Reports assessed for eligibility
(n = 45)
Reports excluded:
Population (n = 9)
Outcomes (n = 17)
Study design (n = 7)
Reports assessed for eligibility
(n = 4)
Reports excluded
(n = 3)
Studies included in review
(n = 16)

- For clinically diagnosed T2D patients (2 studies), rates were 4.6% and 12.5%.
- A study from UK showed that misdiagnoses were higher when insulin treatment was delayed (47%) vs. early insulin treatment (3%).
- LADA misdiagnosed as T2D ranged from 4.2% - 8.3% (median: 5.6%, 4 studies).

Involvement of healthcare professionals

- A French study found no misclassification by diabetologists when diagnosis was compared to the gold standard i.e., C-peptide testing.[21]
- An Australian study revealed that of the 50% of patients diagnosed by general practitioners (GPs), 87% were misdiagnosed as T2D by GPs. GPs were 3.1 times more likely to misdiagnose T1D (95% CI 1.5–6.2) compared to endocrinologists.[15]
- In a UK study, six out of ten LADA patients were diagnosed by GPs or nurses. One patient received a letter from the GP, while three received no specific LADA diagnosis but general diabetes care advice.[7]

Misdiagnosis detection

- Of the nine studies,[7,10,12,16-18,20-22] five identified autoimmune T1D misdiagnosed as T2D, while four reported LADA misdiagnosed as T2D.
- Autoantibody tests such as glutamic acid decarboxylase antibody (GADA), islet tyrosine phosphatase2 (IA2) and zinc transporter 8 (ZNT8) were reported to detect diabetes of autoimmune etiology.[10,12,17,18,20]
- All four LADA studies identified GADA as the most specific marker.[7,16,17,20]

Table 1: Percentage of patients misdiagnosed as T2D

Author & Year	Country	Population	Assessed patients	Misdiagnosis n (%)
Percentage of T1D patients misdiagnosed as T2D				
Cheheltani 2022[13]	USA	Clinically diagnosed T1D	15,881	1,710 (~10)
Hope 2016[9]	UK	Clinically diagnosed T1D	87	30 (34)
Liu 2023[10]	UK	Clinically diagnosed T1D	31	7 (23)
Rodriguez 2023[18]	Spain	Clinically diagnosed T1D	452	205 (45.2)
Takai 2022[19]	Japan	Clinically diagnosed T1D	24	5 (20.8)
Thomas 2019[12]	UK	Clinically diagnosed T1D – Insulin at diagnosis	76	NR (3)
		Clinically diagnosed T1D – Delayed insulin treatment	47	NR (47)
Wright-Pascoe 2000[22]	Jamaica	Clinically diagnosed T2D	8	1 (12.5)
de Lusignan 2012[8]	UK	Clinically diagnosed T2D	241	11 (4.6)
Berkovic 2022[15]	Australia	Self-reported T1D	120	35 (29.2)
Percentage of LADA patients misdiagnosed as T2D				
Davies 2008[7]	UK	Clinically diagnosed T2D	667	28 (4.2)
Manisha 2022[16]	Tanzania	Clinically diagnosed T2D	156	8 (5.1)
Priyadarshini 2021[17]	India	Clinically diagnosed T2D	68	4 (6)
Zaharia 2018[20]	Germany	Clinically diagnosed T2D	NR	NR (8.3)

NR: not reported

- Four studies confirmed C-peptide test accurately distinguishes T1D from T2D.[10,12,17,21] Low fasting C-peptide (<0.6–0.7 ng/ml) or low random C-peptide (<200 pmol/l) strongly suggested T1D, while normal/high levels indicated T2D.
- Two studies highlighted the utility of polygenic risk score/T1D genetic risk score alongside autoantibodies and C-peptide for accurate T1D diagnosis.[10-12]

DISCUSSION

- Misdiagnoses of T1D (22%) and LADA (5.6%) as T2D were lower than global estimates, likely due to demographic differences, denominator variations, and unreported confounders like time to insulin initiation.[2,3]
- The findings of this review align with 2024 ADA guidelines[2] recommending C-peptide and autoantibody tests to differentiate T1D/LADA from T2D.
- The limitations of our review include limited evidence, varied reference populations (T1D, T2D, LADA), and insufficient confounder reporting, making it hard to interpret misdiagnosis rates and causes.
- Most studies had low risk of bias, except for incomplete confounder reporting.

CONCLUSIONS

- Misdiagnoses of T1D (22%) and LADA (5.6%) as T2D is common in clinical practice although estimates varied across populations.
- Specialists such as endocrinologists are less likely to misdiagnose autoimmune T1D as T2D compared to GPs.
- This SLR underscores the importance of enhanced patient and provider education, and accurate diagnosis in early stage via biomarkers (C-peptide, autoantibodies) to delay disease progression and facilitate proper monitoring and treatment in T1D and LADA.

REFERENCES

1. Leslie RD, Evans-Molina C, Freund-Brown J, et al. Adult-Onset Type 1 Diabetes: Current Understanding and Challenges. Diabetes Care. 2021;44(11):2449-2456.
2. American Diabetes Association Professional Practice Committee. 2. Diagnosis and Classification of Diabetes: Standards of Care in Diabetes—2024. Diabetes Care. 2023;47(Supplement_1):S20-S42.
3. Holt RIG, DeVries JH, Hess-Fischl A, et al. The Management of Type 1 Diabetes in Adults. A Consensus Report by the American Diabetes Association (ADA) and the European Association for the Study of Diabetes (EASD). Diabetes Care. 2021;44(11):2589-2625.
4. Higgins JPT, Thomas J, Chandler J, et al., eds. Cochrane Handbook for Systematic Reviews of Interventions. 2nd Edition. Chichester (UK): John Wiley & Sons; 2019.
5. Page MJ, McKenzie JE, Bossuyt PM, et al. The PRISMA 2020 statement: an updated guideline for reporting systematic reviews. International journal of surgery. 2021;88:105906.
6. Moola S, Munn Z, Tufanaru C, et al. Systematic reviews of etiology and risk (2020). In: Aromataris E, Lockwood C, Porritt K, Pilla B, Jordan Z, eds. JBI Manual for Evidence Synthesis.: JBI; 2024.
7. Davies H, Mannan S, Brophy S, Williams R. Routine glutamic acid decarboxylase autoantibody (GADA) testing: patients' perspective. Family practice. 2008;25(3):176-180.
8. de Lusignan S, Sadek N, Mulnier H, Tahir A, Russell-Jones D, Khunti K. Miscoding, misclassification and misdiagnosis of diabetes in primary care. Diabetic medicine : a journal of the British Diabetic Association. 2012;29(2):181-189.
9. Hope SV, Wienand-Barnett S, Shepherd M, et al. Practical Classification Guidelines for Diabetes in patients treated with insulin: a cross-sectional study of the accuracy of diabetes diagnosis. The British journal of general practice : the journal of the Royal College of General Practitioners. 2016;66(646):e315-322.
10. Liu T, Sankareswaran A, Paterson G, et al. Investigating misclassification of type 1 diabetes in a population-based cohort of British Pakistanis and Bangladeshis using polygenic risk scores. medRxiv. 2023:2023.2008.2023.23294497.
11. Seidu S, Davies MJ, Mostafa S, de Lusignan S, Khunti K. Prevalence and characteristics in coding, classification and diagnosis of diabetes in primary care. Postgraduate medical journal. 2014;90(1059):13-17.
12. Thomas NJ, Lynam AL, Hill AV, et al. Type 1 diabetes defined by severe insulin deficiency occurs after 30 years of age and is commonly treated as type 2 diabetes. Diabetologia. 2019;62(7):1167-1172.
13. Cheheltani R, King N, Lee S, et al. Predicting misdiagnosed adult-onset type 1 diabetes using machine learning. Diabetes research and clinical practice. 2022;191:110029.
14. Muñoz C, Floreen A, Garey C, et al. Misdiagnosis and Diabetic Ketoacidosis at Diagnosis of Type 1 Diabetes: Patient and Caregiver Perspectives. Clinical diabetes : a publication of the American Diabetes Association. 2019;37(3):276-281.
15. Berkovic D, Fransquet R, Soh SE, Ayton D. Experiences of adults with adult-onset type 1 diabetes: a cross-sectional study. Australian journal of primary health. 2022;28(4):357-363.
16. Manisha AM, Shangali AR, Mfinanga SG, Mbugi EV. Prevalence and factors associated with latent autoimmune diabetes in adults (LADA): a cross-sectional study. BMC endocrine disorders. 2022;22(1):175.
17. KS Priyadarshini DG, Prashanth Kumar. Glutamic Acid Decarboxylase and Islet Cell Cytoplasmic Autoantibodies Positivity in Latent Autoimmune Diabetes of Adults: Need for Early Insulinisation. Journal of Clinical and Diagnostic Research. 2021;15(5):BC11-BC15.
18. Rodríguez Escobedo R, Lambert C, Morales Sánchez P, Delgado Álvarez E, Menéndez Torre E. Reclassification of type 2 diabetes to type 1 diabetes in Asturias (Spain) between 2011 and 2020. Diabetology & metabolic syndrome. 2023;15(1):90.
19. Takai T, Okada Y, Takebe R, Nakamura T. Vomiting and hyperkalemia are novel clues for emergency room diagnosis of type 1 diabetic ketoacidosis: a retrospective comparison between diabetes types. Diabetology international. 2022;13(1):272-279.
20. Zaharia OP, Bobrov P, Strassburger K, et al. Metabolic Characteristics of Recently Diagnosed Adult-Onset Autoimmune Diabetes Mellitus. The Journal of clinical endocrinology and metabolism. 2018;103(2):429-437.
21. Benhamou PY, Marwah T, Balducci F, et al. Classification of diabetes in patients with end-stage renal disease. Validation of clinical criteria according to fasting plasma C-peptide. Clinical nephrology. 1992;38(5):239-244.
22. Wright-Pascoe R, Mills J, Choo-Kang E, Morrison EY. The role of C-peptide in the classification of diabetes mellitus. The West Indian medical journal. 2000;49(2):138-142.

DISCLOSURES

Laura Wilson and Mariam Hanna are employees of Sanofi**.** Gaurang Nazar, Divya Pushkarna and Boris Breznen are employees of Evidinno Outcomes Research Inc. which was contracted by Sanofi to conduct this research.

FUNDING

Medical writing was provided by Johnny Zhou, an employee, of Evidinno and was funded by Sanofi.