Poster ID 2759030
ISPE 2024
August 24–28
Berlin, Germany

Using Real-World Evidence for Clinical Drug Development to Address the Gap Between Marketing and Authorization and Reimbursement in European Countries

Ramon Hernandez[1], Antoine Pugeat[2], Walid Shouman[3], Divya Pushkarna[3], Jean-Paul Collet[3], Paulo Carita[1]

[1]Sanofi, Campus Gentilly, Paris, France; [2]Sanofi, Campus Sanofi Lyon, Lyon, France; [3]Evidinno Outcomes Research Inc., Vancouver, BC, Canada

To download e-poster scan the QR code

E-poster

INTRODUCTION

- The journey from drug discovery to patient access is a multifaceted process with two crucial milestones, marketing authorization (MA) and pricing & reimbursement (P&R) approval.[1,2]
- Regulatory agencies primarily aim to verify the safety and efficacy of drugs for their intended purposes.
- HTA agencies focus on drug effectiveness, uniqueness, comparative prices, and the severity of condition. Although HTA dossiers differ between countries, there are common requirements across these dossiers that are submitted by manufacturers.[3,4]
- Understanding the role of Real-World Evidence (RWE) to provide relevant clinical evidence and identifying the challenges when submitting RWE would help finding solutions that meet the HTA agencies' requirement; therefore, leading to early acceptance of the drug, which would help reduce the clinical impact of this delay.

OBJECTIVE

To understand the clinical impact and reasons for the gap between MA and P&R of medicines, with a focus on France and Germany, and to explore the potential role of Real-World Data (RWD) in addressing these gaps.

METHODS

- Two Targeted Literature Reviews (TLRs) were conducted:
 - TLR1 focused on the clinical impact and reasons for the gap between MA and P&R of medicines.
 - TLR2 focused on identifying the ways RWE can help address these gaps.
- Both TLRs were conducted following the standard methodologies for conducting and reporting systematic reviews as recommended by the Cochrane Handbook for Systematic Reviews of Interventions.
- Relevant studies for both TLRs were identified by searching Embase using predefined search strategies via the Ovid platform on April 26,2023. Additional records were included from external keyword searches on Google Scholar and PubMed that address the objective of the review.
- Studies reporting on any disease area, intervention, or comparator with outcomes (such as any reported cause of gaps between market authorization and reimbursement of medicines or clinical outcomes due to gap, or outcomes that illustrate the role of RWD in addressing challenges related to the gaps between market authorization and reimbursement of medicines) of interest were included.
- Searches were also conducted in the French Haute Autorité de Santé (HAS) website in France and the Gemeinsamer Bundesausschuss (GBA) in Germany to identify reimbursement submissions in which RWE has played a role in achieving a positive decision.

RESULTS

- TLR1 included guidelines and systematic reviews (n = 4 each), retrospective database searches and webpages (n = 3 each), commentary papers, and surveys (n = 2 each), and book chapter, analytical framework, annual report, and multi-year annual metrics study (n = 1 each).
- TLR2 included descriptive reviews (n = 21), followed by webpages (n = 5) and cohort study, systematic literature review and a template for planning and reporting (n = 1 each).

Figure 1: PRISMA diagram (TLR1 - Impact of gap between MA and P&R; TLR2 - (Use of RWE to address gaps between MA and P&R)

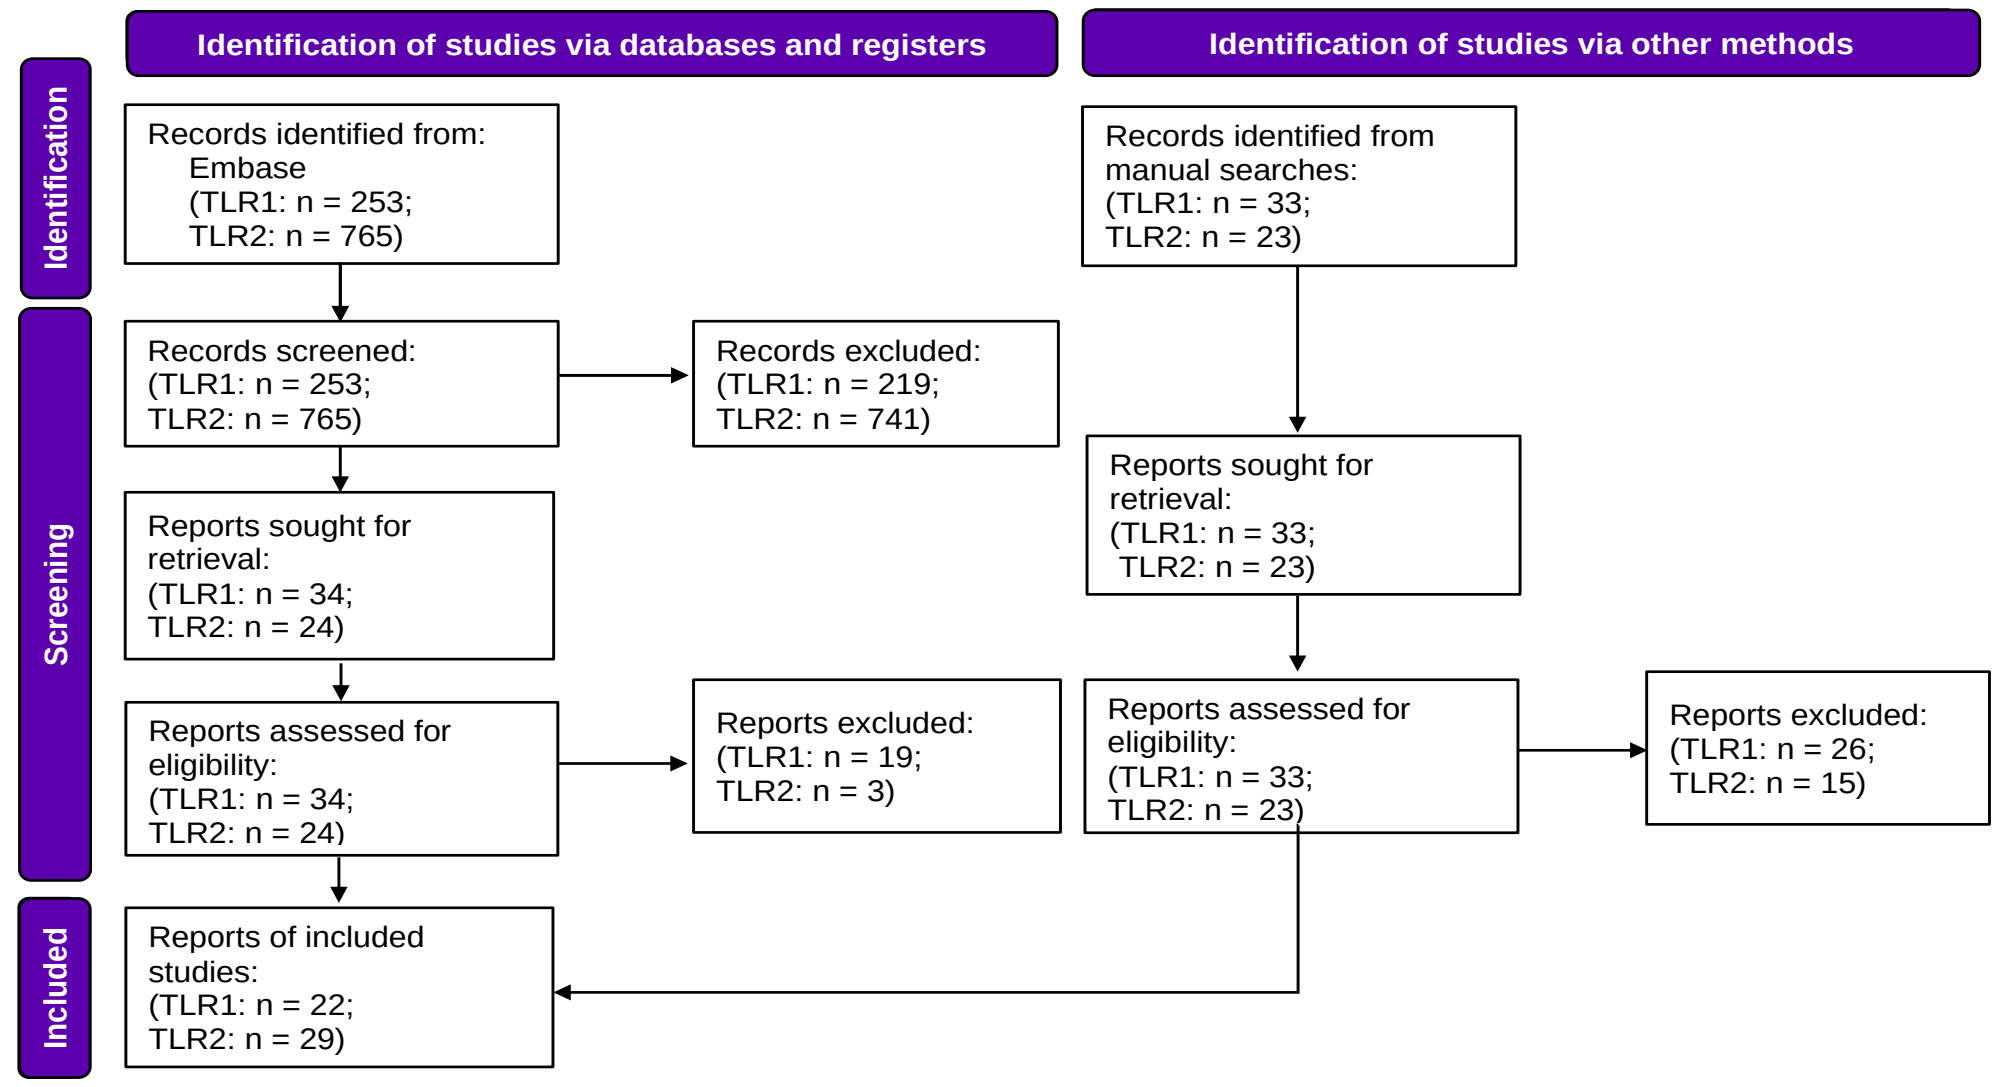
Identification of studies via databases and registers
Identification of studies via other methods
Identification
Screening
Included
Records identified from: Embase (TLR1: n = 253; TLR2: n = 765)
Records identified from manual searches: (TLR1: n = 33; TLR2: n = 23)
Records screened: (TLR1: n = 253; TLR2: n = 765)
Records excluded: (TLR1: n = 219; TLR2: n = 741)
Reports sought for retrieval: (TLR1: n = 34; TLR2: n = 24)
Reports sought for retrieval: (TLR1: n = 33; TLR2: n = 23)
Reports assessed for eligibility: (TLR1: n = 34; TLR2: n = 24)
Reports excluded: (TLR1: n = 19; TLR2: n = 3)
Reports assessed for eligibility: (TLR1: n = 33; TLR2: n = 23)
Reports excluded: (TLR1: n = 26; TLR2: n = 15)
Reports of included studies: (TLR1: n = 22; TLR2: n = 29)

Documenting the delay between market authorization and drug reimbursement

- Patient Waiting to Access Innovative Therapies (W.A.I.T.) indicator 2022 survey highlighted that the average time to reimbursement for innovative treatments across Europe was on average 517 days with variations from 128 days in Germany and 508 days in France.
- The expected time to receive an HTA approval varies widely among countries. Spain, for instance, contends with a protracted reimbursement timeline of approximately 629 days, a reflection of the involvement of multiple agencies in the evaluation process.[5]
- Delay of access to treatment in prostate cancer patients eligible for abiraterone resulted in 55,853 non-treated patients, which would indicate a loss of 18,152 life-years across Europe for abiraterone non-use.[6]
- Delay in HTA approval is majorly due to the lack of evidence that support the added value of the drug. Hence, HTA bodies end up requesting additional data during the approval process.
- Further reasons include the lack of safety and efficacy data from the submitted clinical trials and the absence of direct comparative data versus clinically relevant comparators.
- More recently, the lack of comparative data is being solved by using RWD/RWE following access to reimbursement.

Figure 3: Overview of the reasons of delay in achieving HTA approvals. [7-12]

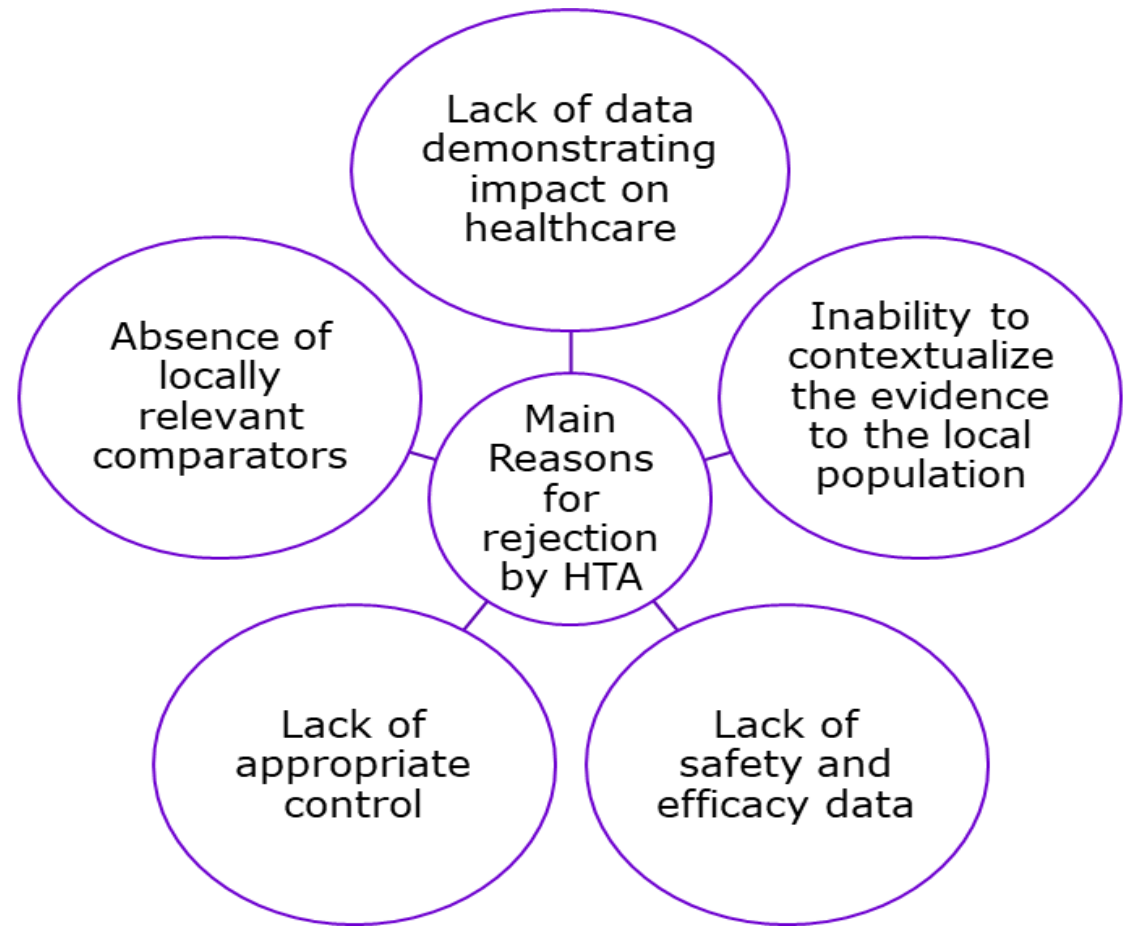
Lack of data demonstrating impact on healthcare
Absence of locally relevant comparators
Main Reasons for rejection by HTA
Inability to contextualize the evidence to the local population
Lack of appropriate control
Lack of safety and efficacy data

The use of clinical RWE to address the delay

- Recently, there has been a growing emphasis on incorporating clinical RWE into the decision-making processes of P&R. RWE has shown to help address the reasons for delay in achieving HTA approval, especially due to lack of appropriate clinical data.
- Data from Electronic Health Records (EHRs) can help address various safety issues, especially long-term safety data that are often not detected over a limited duration phase III trial.[13,14] Moreover, RWD can serve as a source of external control in single arm trials.
- RWE can also be leveraged in addition to RCTs to increase the completeness of evidence-based medicine generated for clinical prescription guidelines.

Figure 4: Ways RWE can be utilized in HTA submissions

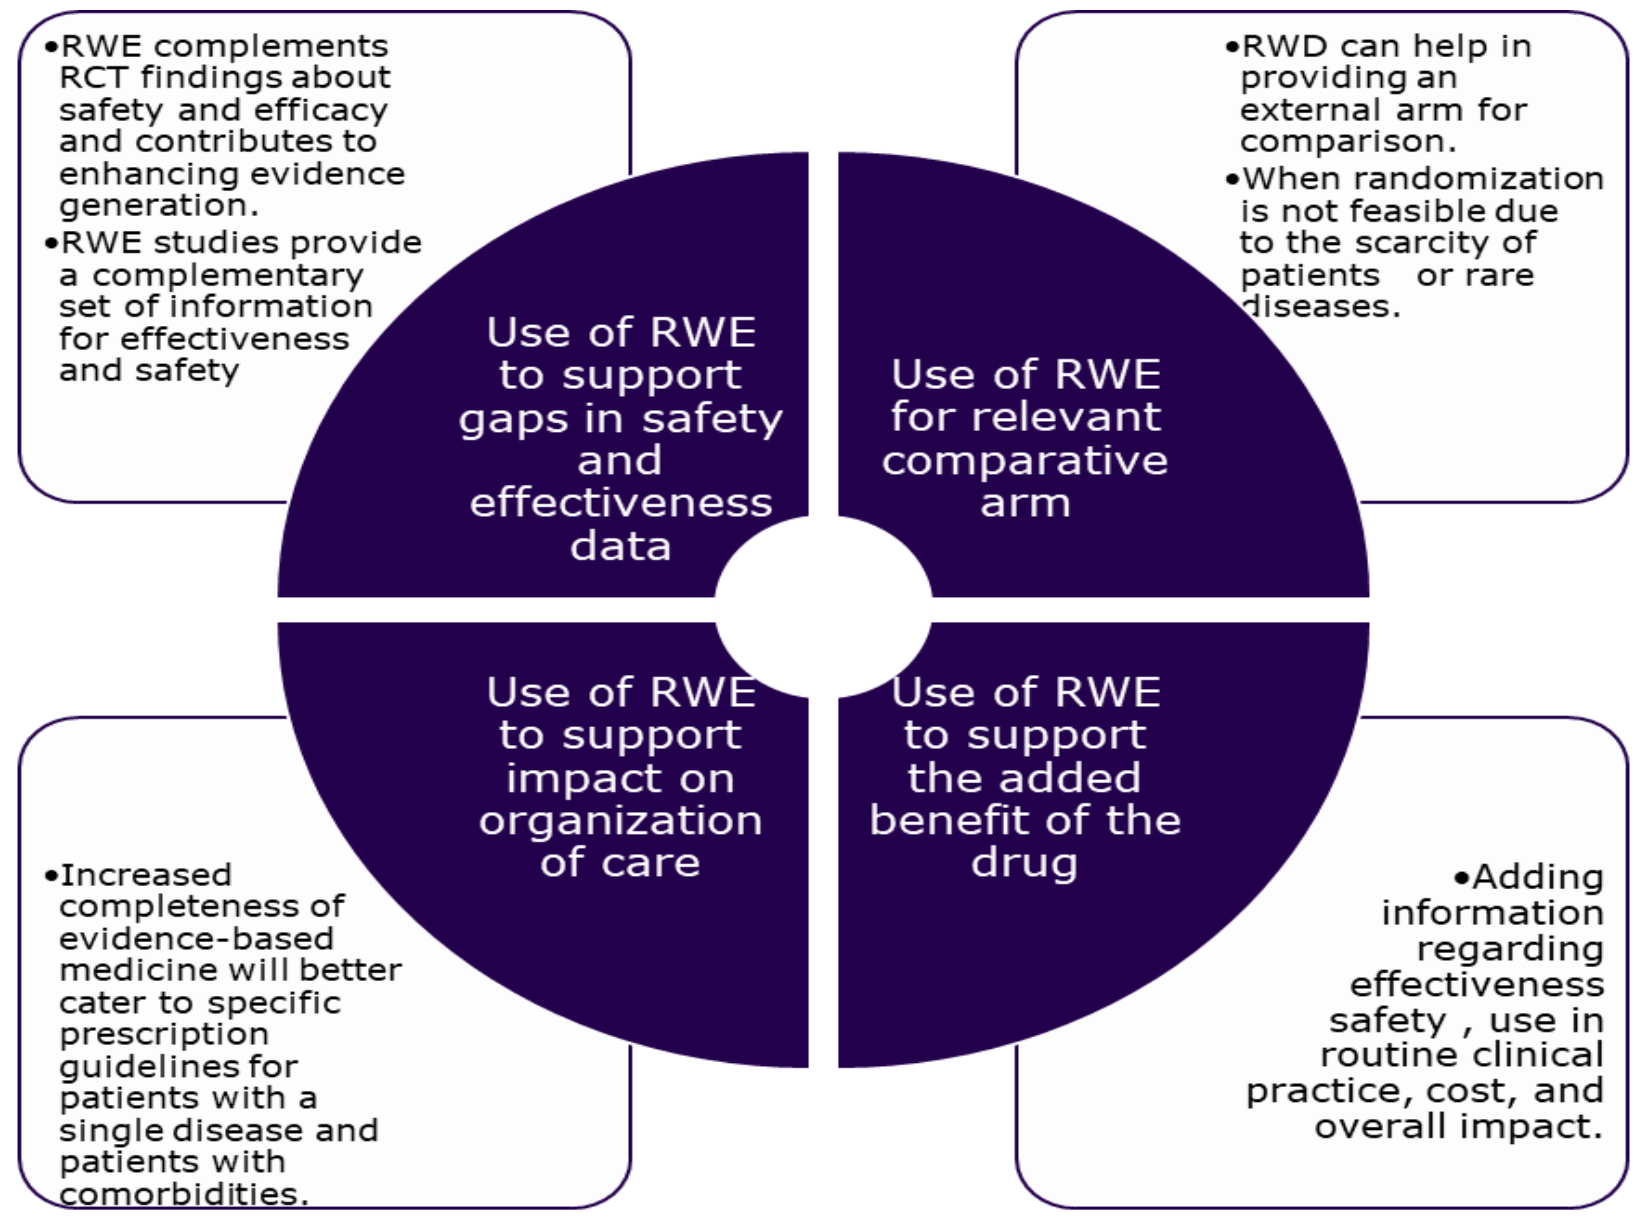
•RWE complements RCT findings about safety and efficacy and contributes to enhancing evidence generation.
•RWE studies provide a complementary set of information for effectiveness and safety
•RWD can help in providing an external arm for comparison.
•When randomization is not feasible due to the scarcity of patients or rare diseases.
Use of RWE to support gaps in safety and effectiveness data
Use of RWE for relevant comparative arm
Use of RWE to support impact on organization of care
Use of RWE to support the added benefit of the drug
•Increased completeness of evidence-based medicine will better cater to specific prescription guidelines for patients with a single disease and patients with comorbidities.
•Adding information regarding effectiveness safety , use in routine clinical practice, cost, and overall impact.

DISCUSSION

- In practice, there are some limitations for the use of RWE. These limitations include poor data quality, unprecise research questions,[17] limited scientific approaches,[18] and issues with transparency and credibility.
- In addition, since the variation in evidentiary requirements is a cause of delay to market access, harmonization is essential to avoid duplicative efforts in post-launch evidence generation.
- Traditional clinical trials have limitations, like strict inclusion criteria and controlled settings, which may not reflect real-world populations and outcomes. RWE/RWD address this by offering broader insights into treatment outcomes, patient preferences, and long-term safety.
- RWE offers robust evidence for cost-effectiveness analyses, helping stakeholders evaluate interventions' added value and align pricing with real-world outcomes.
- A remaining challenge is the differing perceptions of epidemiologic data versus randomized clinical trials among HTA agencies, along with their specific requirements for determining a new drug's clinical and economic "added value." Harmonizing HTA review processes across countries will take more time.

CONCLUSIONS

- Real-World Evidence (RWE) and Real-World Data (RWD) are transforming healthcare by reducing delays in medication access caused by HTA processes, filling knowledge gaps, and facilitating patient-centered approaches.
- The effective use and collaboration among stakeholders in RWE/RWD are crucial for informed and equitable healthcare reimbursement decisions and pricing structures.

REFERENCES

1. Ferrario A, et al. Value in Health. 2018;21(7):809-821.
2. Wilking N, et al. Karolinska Institutet Stockholm, Sweden; 2005.
3. White P, et al. 2023.
4. Chitkara A, et al. Journal of Clinical Oncology. 2023.
5. Newton M, et al. EFPIA Patients WAIT Indicator;2022.
6. Uyl-de Groot, et al. Cancers. 2020;12(8):2313.
7. Mann H, et al. Journal of the Royal Society of Medicine. 2013;106(1):30-33.
8. Malay S, et al. Plastic and reconstructive surgery. 2012;130(4):959.
9. Wang T, et al. Frontiers in pharmacology. 2020;11:594549.
10. de Pouvourville G, et al. Value in Health. 2023;26(4):3-10.
11. Jpt H, et al. http://www cochrane-handbook org. 2008.
12. Fogel DB. et al. 2018;11:156-164.
13. Dang A. et al. Pharmaceutical Medicine. 2023;37(1):25-36.
14. Viera AJ, et al. FAMILY MEDICINE-KANSAS CITY-. 2007;39(2):132.
15. Dang A, et al. International Journal of Risk & Safety in Medicine. 2021;32(3):163-173.
16. Hiramatsu K, et al. Drugs-Real World Outcomes. 2021;8(4):459-480.
17. Reynolds MW, et al. Pharmacoepidemiology and drug safety. 2020;29(10):1316.
18. Liu M, et al. European Journal of Hospital Pharmacy. 2022;29(1):8-11.

DISCLOSURES:

JC, DP, and WS report employment with Evidinno Outcomes Research Inc. RH, AP, and PC report employment with Sanofi. Authors report no other conflicts of interest.

FUNDING:

The study was sponsored by Sanofi and conducted by Evidinno Outcomes Research Inc.