222

Can overall survival benefit be predicted from improvements in progression-free survival (PFS) for previously untreated metastatic colorectal cancer?

Jeanine ML Roodhart[1], Kiran Dave[2], Matthew Dixon[3], Murat Kurt[3], Divya Pushkarna[4], Mir-Masoud Pourrahmat[4], Mir Sohail Fazeli[4]

[1]University Medical Center, Utrecht, Netherlands; [2]Bristol Myers Squibb, Uxbridge, UK; [3]Bristol Myers Squibb, Princeton, NJ, USA; [4]Evidinno Outcomes Research Inc., Vancouver, BC, Canada

Introduction

- Colorectal cancer (CRC) is the third most common cancer worldwide, accounting for 10% of all diagnoses.[1] Among metastatic CRC patients, the 5-year survival rate is only 14%,[2] highlighting the need for better treatments.
- Overall survival (OS) is the standard measure of efficacy in oncology, but obtaining mature OS data can be challenging in cases involving effective treatments or populations exhibiting specific biomarkers (e.g., PD-L1 expression, microsatellite instability) by requiring extended follow-up durations and larger trials.[3,4]
- To address these challenges, surrogate endpoints such as progression-free survival (PFS) are often used as primary endpoints in randomized settings. In contrast to OS, PFS allows earlier efficacy assessment, higher statistical power with fewer patients, and avoids the influence of post-progression therapy on efficacy assessment.[5,6]
- Elston and Taylor (2009) proposed three criteria for validating surrogates: aggregate-level association, individual-level association, and biological plausibility.[7]
- Building on prior evidence of individual-level association between PFS and OS, this study aimed to address the association criteria at the trial level by evaluating the correlation between treatment effects on PFS and OS. based on aggregate-level data from RCTs in first-line metastatic CRC (mCRC).[8]

Methods

Systematic Literature Review

- Embase, MEDLINE®, and Cochrane Controlled Register of Trials (CENTRAL) were searched from inception to July 2021. Conference proceedings (2019–2021) and US/EU clinical trial registries were also reviewed.
- Eligible studies included randomized (phase 2-4) and non-randomized clinical trials of adults (≥18 years) with 1L mCRC, reporting hazard ratios (HRs) for intervention vs. comparator or Kaplan-Meier (KM) curves by arm for PFS and OS.

Statistical Methods

- The correlation between HR_{PFS} and HR_{OS} was analyzed on a natural log scale using a revised bivariate random-effects meta-analysis (BRMA) and weighted linear regression (WLR).
- Predictive performance was evaluated with leave-one-out cross-validation (LOOCV), assessing if the 95% prediction interval (PI) captured observed HR_{OS} as a validity measure.
- Surrogate threshold effects (STE), the minimum PFS benefit translating to OS benefit with 95% probability, were derived for various sample sizes to gauge model utility in prospective RCTs.
- The primary analysis consisted of all included trials in the evidence base. Sensitivity analyses omitted trials that 1) had anti-EGFR medications, 2) had anti-VEGF medications (due to differences in their mechanisms of action), 3) violated the proportional hazards (PH) assumption of the Cox PH model, to assess the impact of these studies on the analysis, and 4) permitted treatment crossover, to assess the impact of treatment crossover on the results.

Results

Systematic Literature Review

- The SLR identified 178 trials, of these, 47 studies published in 2010 or later with comparators limited to chemotherapy alone or chemotherapy + anti-VEGF/anti-EGFR were included in the analyses. Sample sizes ranged from 48 to 3,058 with a total of 13,959 patients.
- In the evidence base, there were 1 phase I/II, 27 phase II, 1 phase II/III, 16 phase III and 1 phase IV trial. One trial did not report its phase. Geographically, 18 trials were multinational, 14 in Europe, 5 each in Japan and the US, 3 in China, and 1 each in Canada and Algeria.

Primary analysis

- The surrogacy equation derived from WLR was $\log(HR_{OS}) = -0.03 + 0.56 \log(HR_{PFS})$ with a statistically insignificant intercept and statistically significant slope emphasizing the strength of the relationship between HR_{PFS} and HR_{OS} (**Figure 1**).
- The estimated correlation between HR_{PFS} and HR_{OS} was 0.67 (95% CI: 0.48–0.80) using BRMA and 0.70 (95% CI: 0.48–0.84) using WLR (**Table 1**).
- For hypothetical trials with 200 and 300 patients, WLR estimated that PFS HRs less than 0.55 and 0.62, respectively, could lead to statistically significant OS benefit ($HR_{OS} < 1$) at the 95% confidence level.

Figure 1: Scatterplot from weighted linear regression.

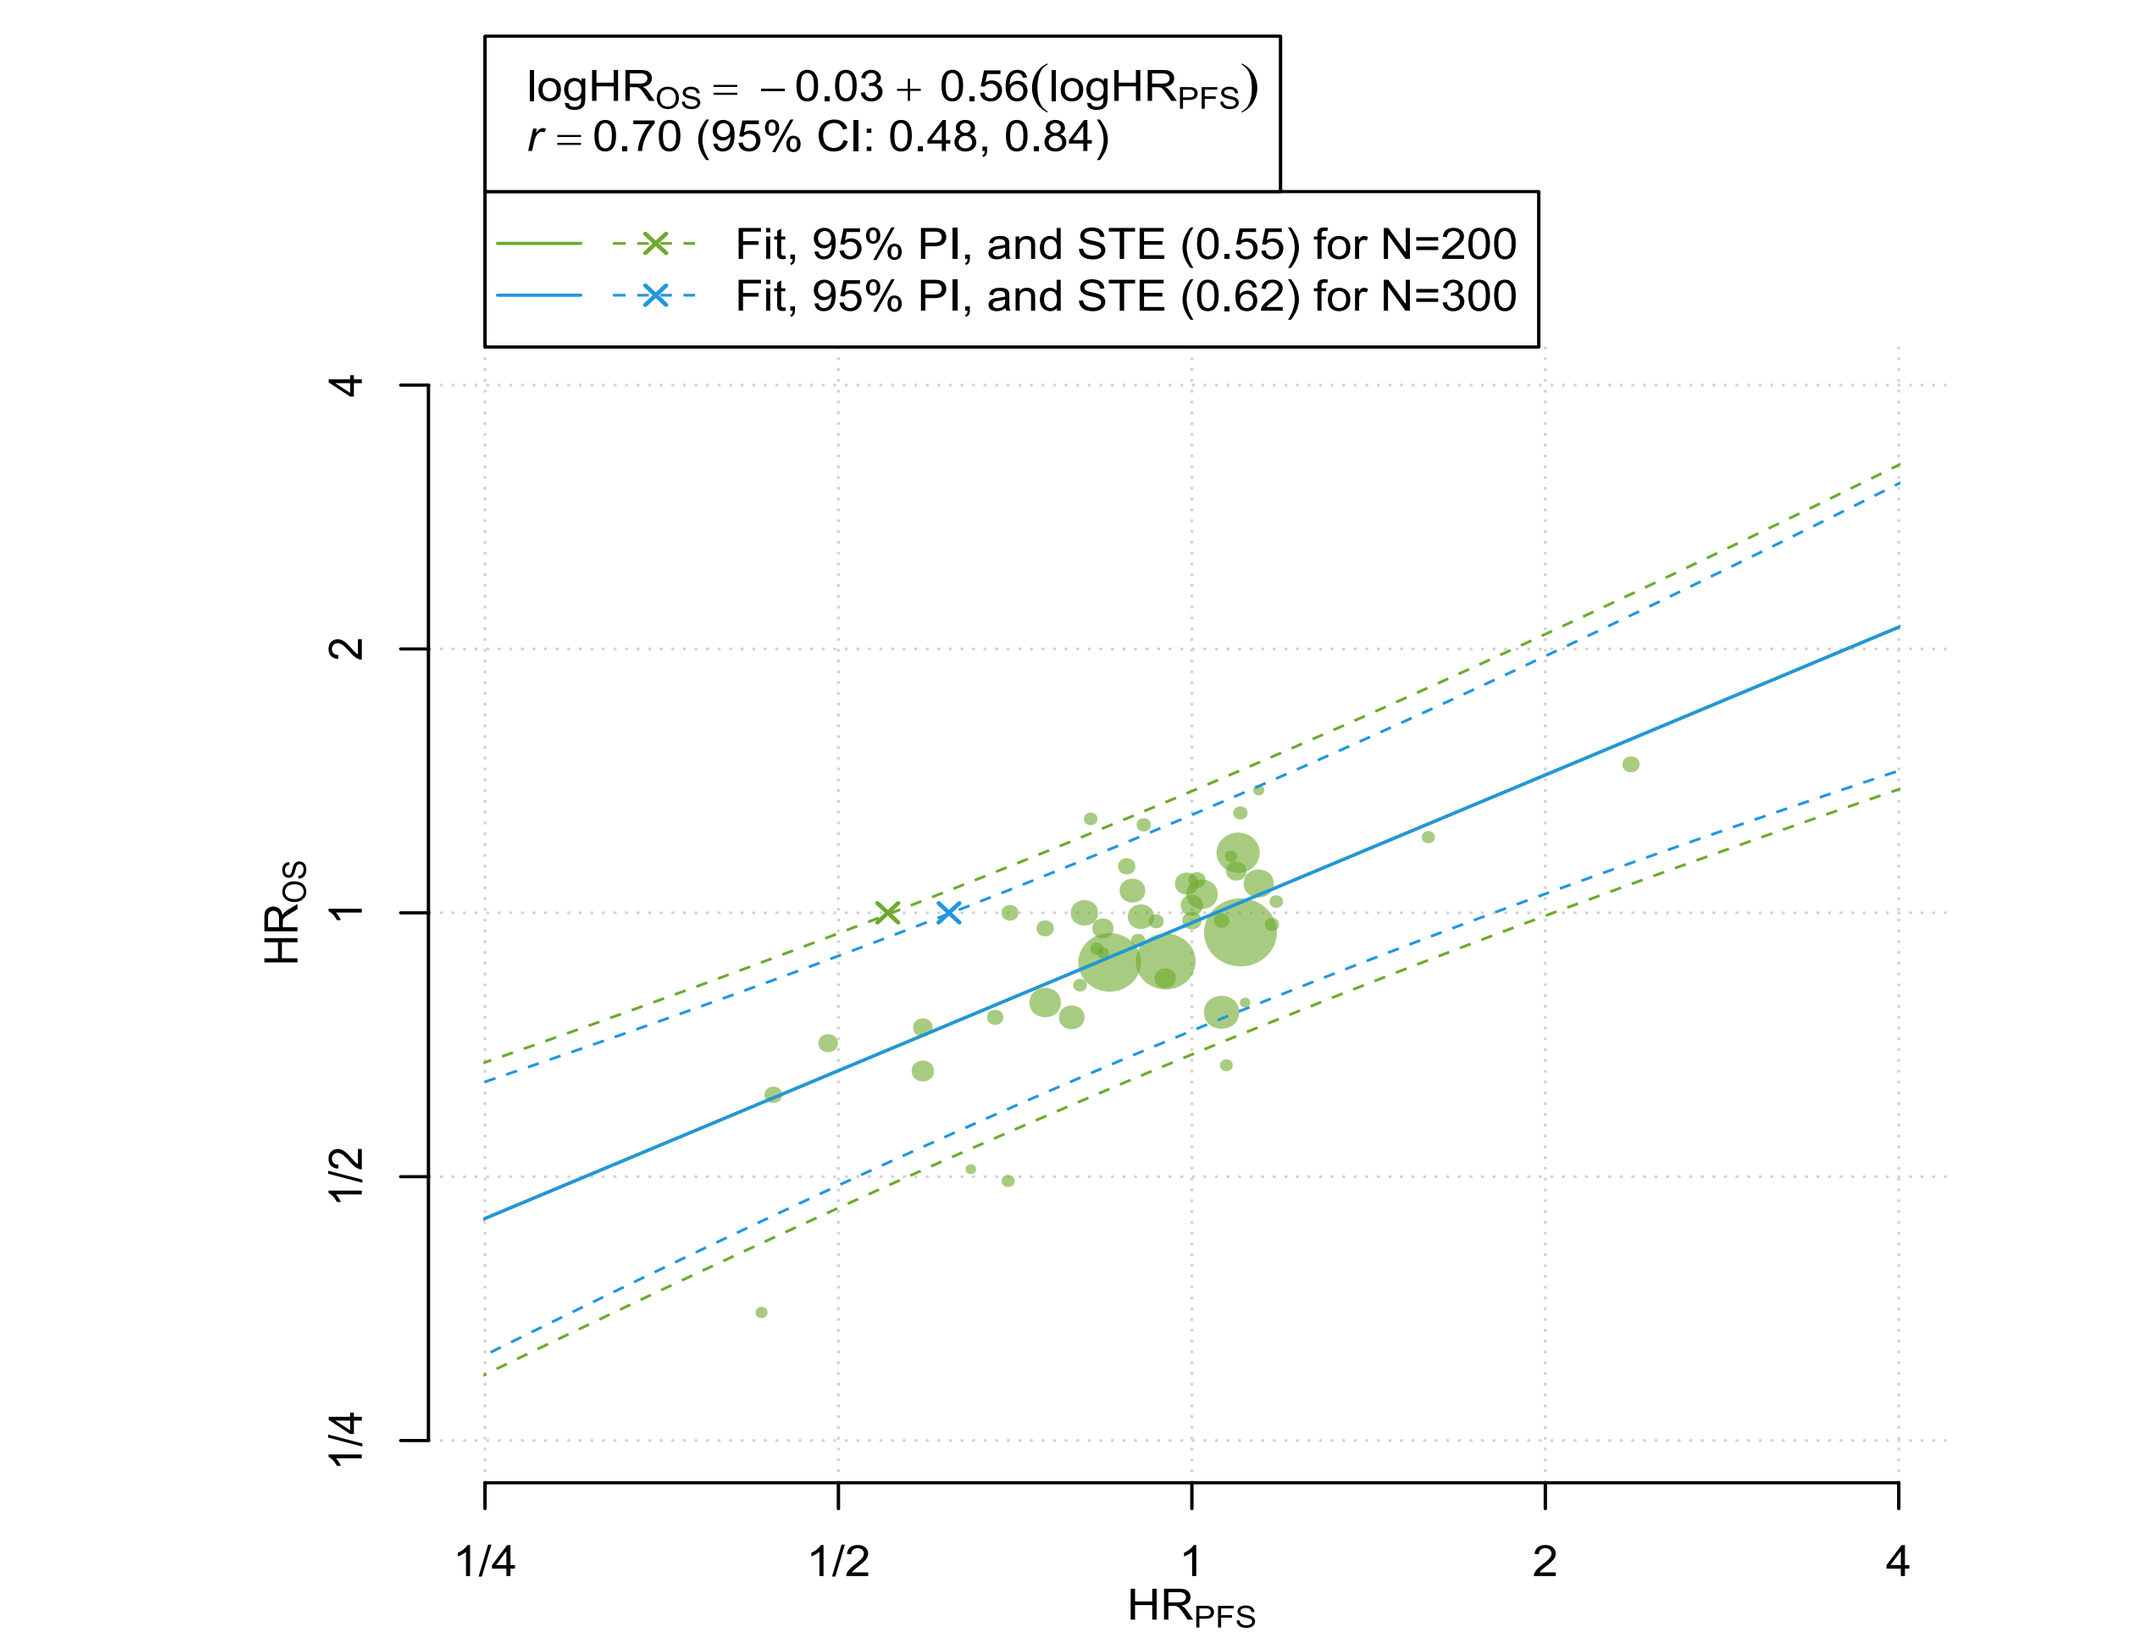
logHR_OS = −0.03 + 0.56(logHR_PFS)
r = 0.70 (95% CI: 0.48, 0.84)
Fit, 95% PI, and STE (0.55) for N=200
Fit, 95% PI, and STE (0.62) for N=300
HR_OS
4
2
1
1/2
1/4
1/4
1/2
1
2
4
HR_PFS

__Legend__: The WLR is graphed as a solid straight line (—) with its corresponding 95% predictive interval boundaries as dotted curved lines (---). The green dots (●) are plotted using measures from the reported $\log(HR_{PFS})$ on the x-axis against measures of $\log(HR_{OS})$ on y-axis for each treatment comparison. Sizes of the dots correspond to the weights associated within the surrogacy equation. Abbreviations: HR – Hazard ratio; OS – Overall survival; PFS – Progression-free survival.

Table 1: Correlation, STE, and LOOCV results.

Analysis Set	# of Studies	Correlation (95% CI)		STE		LOOCV Coverage Rate
		BRMA	WLR	N = 200	N = 300	
Primary Analysis	47	0.67 (0.48, 0.80)	0.70 (0.48, 0.84)	0.55	0.62	93.6%
SA 1 – Anti-EGFR	34	0.75 (0.57, 0.86)	0.78 (0.55, 0.90)	0.55	0.61	94.1%
SA 2 – Anti-VEGF	14	0.44 (-0.08, 0.77)	0.62 (-0.05, 0.90)	0.44	0.52	92.9%
SA 3 – Proportional Hazards	36	0.61 (0.36, 0.77)	0.59 (0.26, 0.80)	0.46	0.55	94.4%
SA 4 - Crossover	43	0.68 (0.48, 0.81)	0.67 (0.41, 0.83)	0.52	0.60	93.0%

CI – confidence interval, LOOCV – leave-one-out cross validation, N – sample size, SA – sensitivity analysis, STE – surrogate threshold effect.

Figure 2: WLR model on treatment comparisons developed for treatment-level correlation.

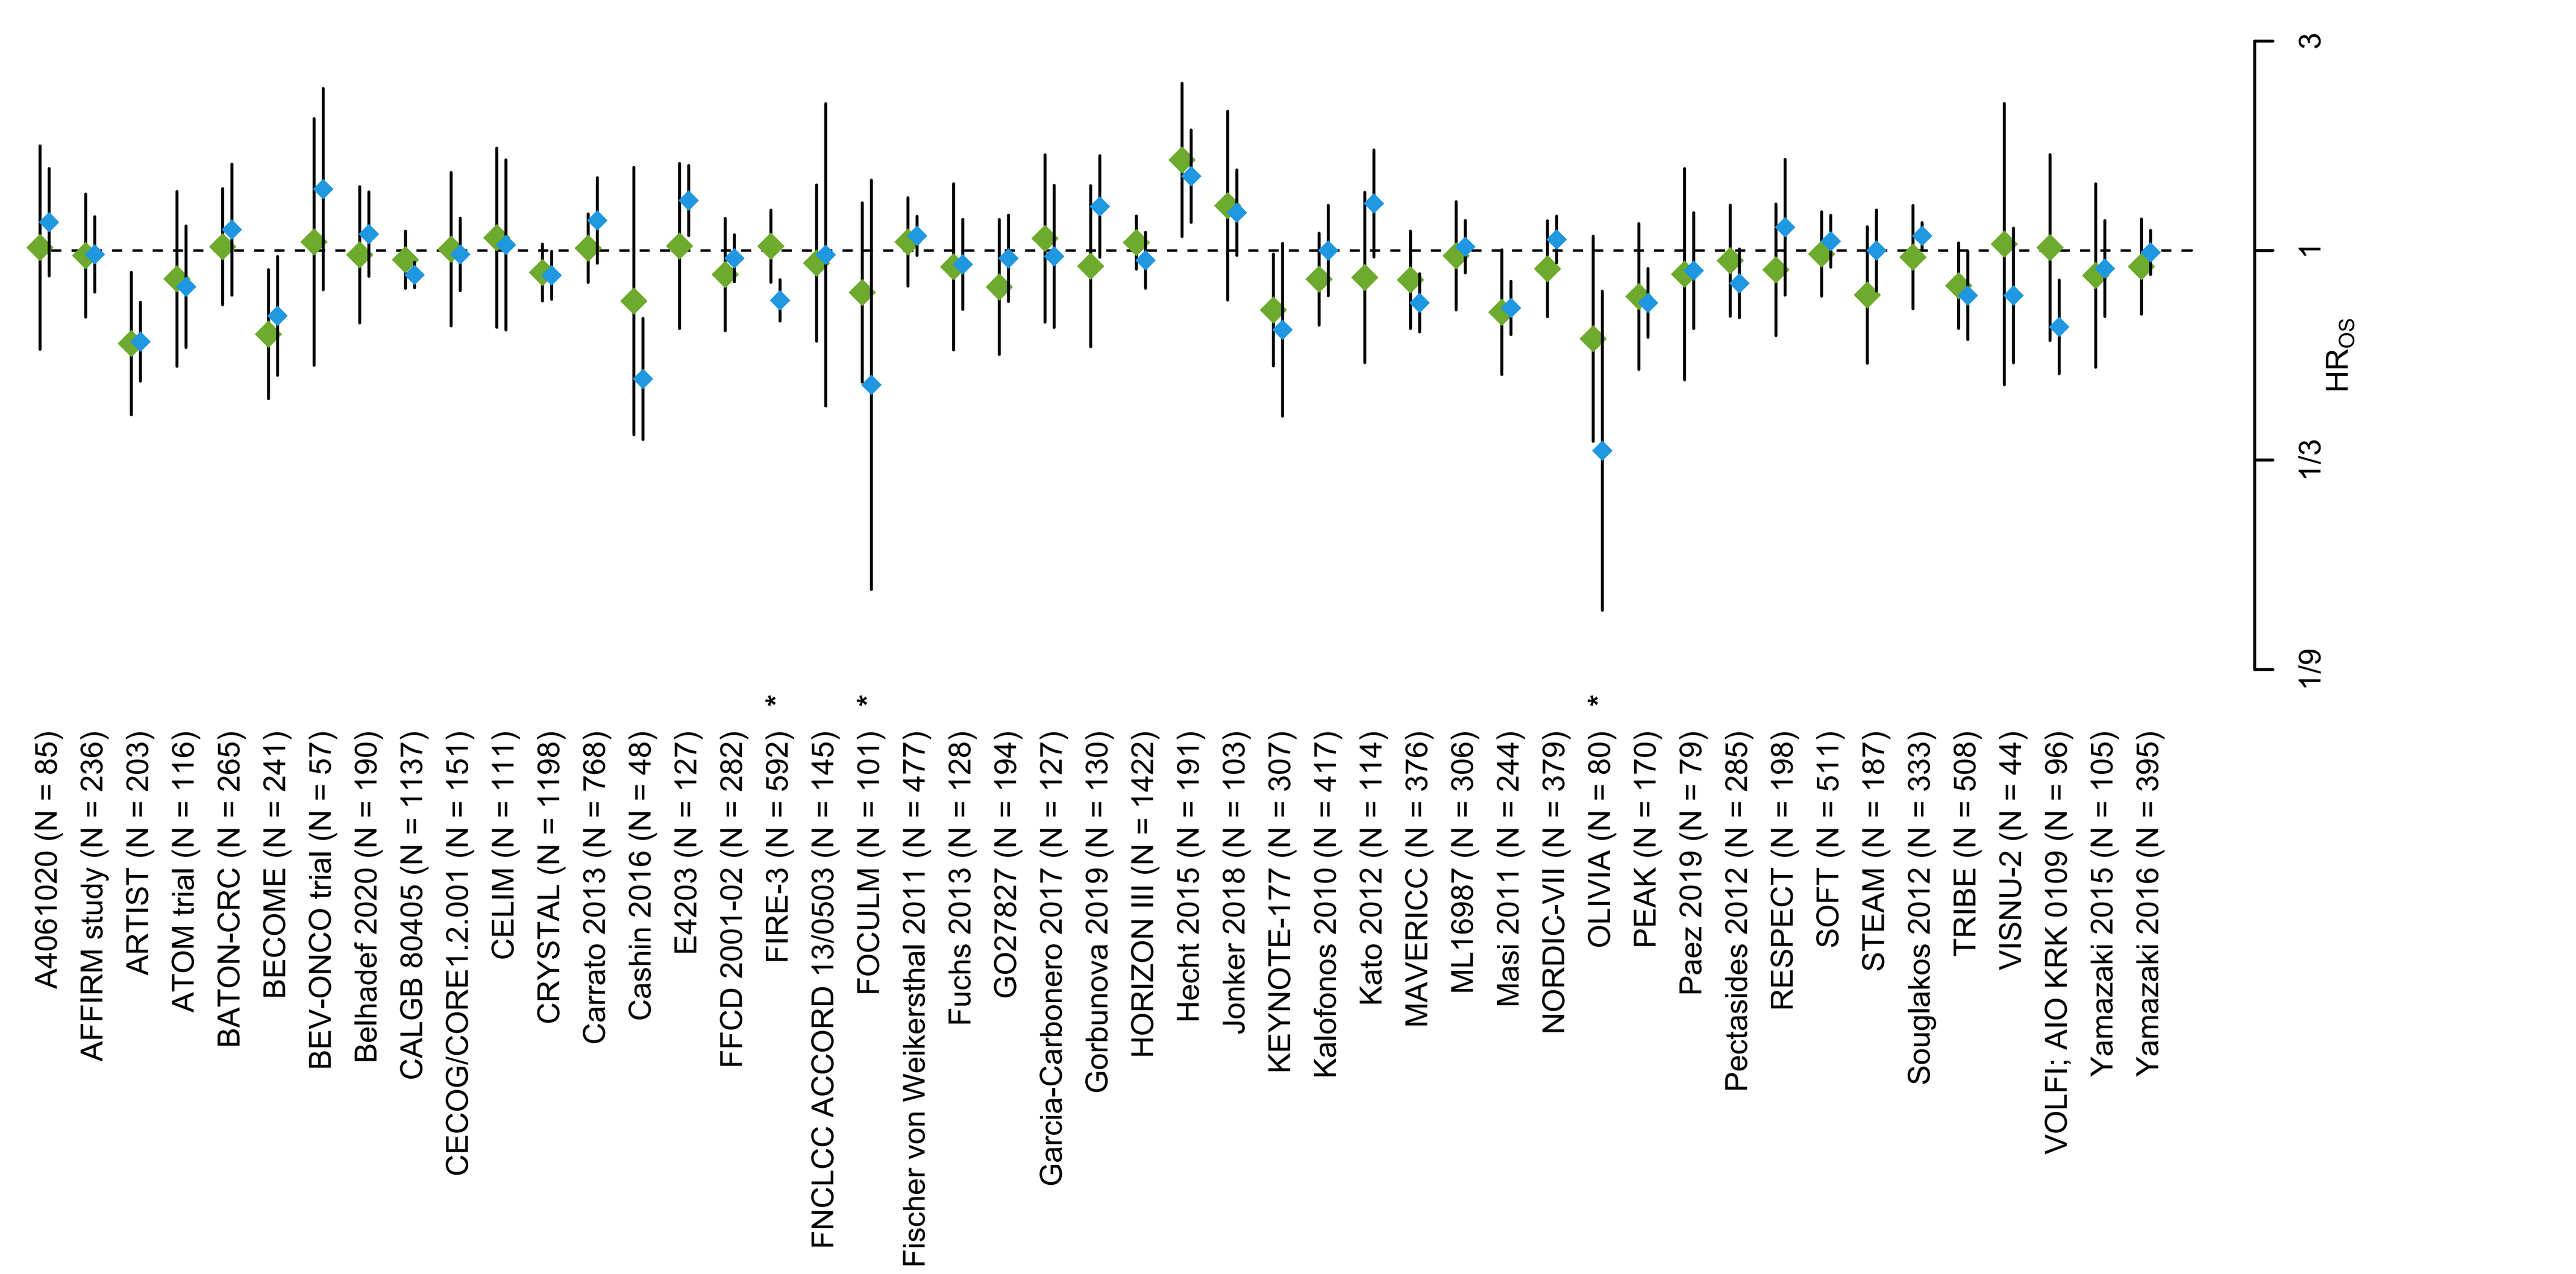
A4061020 (N = 85)
AFFIRM study (N = 236)
ARTIST (N = 203)
ATOM trial (N = 116)
BATON-CRC (N = 265)
BECOME (N = 241)
BEV-ONCO trial (N = 57)
Belhadef 2020 (N = 190)
CALGB 80405 (N = 1137)
CECOG/CORE1.2.001 (N = 151)
CELIM (N = 111)
CRYSTAL (N = 1198)
Carrato 2013 (N = 768)
Cashin 2016 (N = 48)
E4203 (N = 127)
FFCD 2001-02 (N = 282)
FIRE-3 (N = 592) *
FNCLCC ACCORD 13/0503 (N = 145)
FOCULM (N = 101) *
Fischer von Weikersthal 2011 (N = 477)
Fuchs 2013 (N = 128)
GO27827 (N = 194)
Garcia-Carbonero 2017 (N = 127)
Gorbunova 2019 (N = 130)
HORIZON III (N = 1422)
Hecht 2015 (N = 191)
Jonker 2018 (N = 103)
KEYNOTE-177 (N = 307)
Kalofonos 2010 (N = 417)
Kato 2012 (N = 114)
MAVERICC (N = 376)
ML16987 (N = 306)
Masi 2011 (N = 244)
NORDIC-VII (N = 379)
OLIVIA (N = 80) *
PEAK (N = 170)
Paez 2019 (N = 79)
Pectasides 2012 (N = 285)
RESPECT (N = 198)
SOFT (N = 511)
STEAM (N = 187)
Souglakos 2012 (N = 333)
TRIBE (N = 508)
VISNU-2 (N = 44)
VOLFI; AIO KRK 0109 (N = 96)
Yamazaki 2015 (N = 105)
Yamazaki 2016 (N = 395)
HR_OS
3
1
1/3
1/9

__Legend__: The blue diamond (♦) and its error bars are the observed HR_{OS} and its 95% confidence interval, the green diamond (♦) and its error bars are the predicted HR_{OS} and its 95% prediction interval, and an asterisk in the margin indicates that the observed HR_{OS} is not captured by the 95% prediction interval. Abbreviations: HR – Hazard ratio; OS – Overall survival.

Results

- Observed HR_{OS}'s were within their 95% prediction intervals predicted from HR_{PFS} for 93.6% of studies in LOOCV (**Figure 2**).

Sensitivity analyses

- Sensitivity analyses resulted in moderate correlations and slightly lower STEs than the primary analysis and maintained >90% alignment between the observed HR_{OS} and 95% PIs of the predicted HR_{OS}'s from the model in LOOCV.
- Excluding trials of anti-EGFR therapies and trials that permitted treatment crossover improved the strength of association but omitting studies with anti-VEGF medications and studies which violated the PH assumption reduced the correlation.

Discussion

- The analysis results aligned with Shi et al. (2015),[9] showing a slightly higher correlation of 0.73, and exceeded Yoshida et al. (2020),[10] which reported a weaker correlation of 0.57; differences in study selection and Yoshida's use of unweighted Spearman's rho may explain this disparity.
- The correlation strength matches typical oncology surrogacy analyses; among 193 trial-level correlations for OS, 18% were ≥0.85, 28% fell between 0.70 and 0.85, and 55% were <0.70.[11]
- However, these findings should be cautiously interpreted, as STEs are largely based on chemotherapy and targeted therapy trials, limiting applicability to newer therapies such as immune-checkpoint inhibitors in MSI-H and MMR-deficient tumors.

Conclusions

- Moderate correlations between HR_{PFS} and HR_{OS} using BRMA and WLR demonstrate consistent findings across methodologies.
- Cross-validation of surrogacy equations suggests PFS benefit can predict OS benefit in previously untreated mCRC.
- These findings validate PFS as a surrogate endpoint for OS but require further verification with new therapies and specific biomarker subgroups like MSI-H and MMR.

References

1. Sung H, Ferlay J, Siegel RL, et al. Global Cancer Statistics 2020: GLOBOCAN Estimates of Incidence and Mortality Worldwide for 36 Cancers in 185 Countries. CA: a cancer journal for clinicians. May 2021;71(3):209-249. doi:10.3322/caac.21660
2. ACS. Cancer Facts & Figures 2021. 2021
3. Lux MP, Ciani O, Dunlop WCN, et al. The Impasse on Overall Survival in Oncology Reimbursement Decision-Making: How Can We Resolve This? Cancer management and research. 2021;13:8457-8471.
4. Pazdur R. Endpoints for assessing drug activity in clinical trials. Oncologist. 2008;13 Suppl 2:19-21.
5. Han K, Ren M, Wick W, et al. Progression-free survival as a surrogate endpoint for overall survival in glioblastoma: a literature-based meta-analysis from 91 trials. Neuro-oncology. 2014;16(5):696-706.
6. FDA. Clinical Trial Endpoints for the Approval of Cancer Drugs and Biologics Guidance for Industry. December 2018. https://www.fda.gov/media/71195/download
7. Taylor RS, Elston J. The use of surrogate outcomes in model-based cost-effectiveness analyses: a survey of UK Health Technology Assessment reports. International Journal of Technology Assessment in Health Care. 2009;13(8):6-13.
8. Roodhart, JML., Sharpe, DJ, Tate, AE, Bertwistle, D, Dixon, M, Chen, HH., Kurt, M. 57P Evaluating the patient-level association between progression-free survival and overall survival in microsatellite instability-high/mismatch repair-deficient metastatic colorectal cancer (MSI-H/dMMR mCRC) treated with immune checkpoint inhibitors. 2024;35;S27.
9. Qian Shi et al., Individual Patient Data Analysis of Progression-Free Survival Versus Overall Survival As a First-Line End Point for Metastatic Colorectal Cancer in Modern Randomized Trials: Findings From the Analysis and Research in Cancers of the Digestive System Database. JCO 33, 22-28(2015).
10. Yoshida, Y., Kaneko, M. & Narukawa, M. Magnitude of advantage in tumor response contributes to a better correlation between treatment effects on overall survival and progression-free survival: a literature-based meta-analysis of clinical trials in patients with metastatic colorectal cancer. Int J Clin Oncol 25, 851–860 (2020).
11. Haslam A, Hey SP, Gill J, Prasad V. A systematic review of trial-level meta-analyses measuring the strength of association between surrogate end-points and overall survival in oncology. European Journal of Cancer. 2019;106:196-211. doi:10.1016/j.ejca.2018.11.012

Acknowledgments

- All authors contributed to and approved the presentation; writing assistance was provided by Evidinno Outcomes Research Inc., funded by Bristol Myers Squibb.

Presented at the Gastrointestinal Cancers Symposium (ASCO-GI); January 23-25, 2025; San Francisco, CA and Virtual

Contact: mfazeli@evidinno.com

This poster may not be reproduced without permission from ASCO® or the author of this poster.