Literature Review of Safety Outcomes in Chronic Lymphocytic Leukemia (CLL): First-line (1L) Fixed Duration and Monotherapy Treatment Options in Canada

Vuong, V.[1], Kasireddy, E.[2], May, J.R.[2], Collet, J.[2], Pourrahmat, M.[2], Fazeli, M.S.[2], Dutta, N.[1], Kim, C.[1]

[1]AstraZeneca, Mississauga, Canada; [2]Evidinno Outcomes Research Inc

Background

- Chronic lymphocytic leukemia (CLL) is the most common type of leukemia in Canada[1], with an estimated 1,700 new cases in 2019 and 555 deaths in 2022.[2-4]
- CLL is a slowly progressing, chronic disease with a 5-year net survival rate of 86%[3], leading to a significant burden for patients and the healthcare system.[5]
- Currently available first-line (1L) treatment options include[6,7]:
 - Targeted treat-to-progression (TTP) monotherapy with a Bruton's tyrosine kinase inhibitor (BTKi) (acalabrutinib, ibrutinib, and zanubrutinib).
 - Targeted fixed-duration combinations (ibrutinib + venetoclax [**I+V**] and venetoclax + obinutuzumab [**V+O**]).
 - Chemoimmunotherapy (e.g., fludarabine-cyclophosphamide-rituximab [FCR], bendamustine-rituximab [BR], and chlorambucil + obinutuzumab).
- A novel targeted fixed-duration regimen, acalabrutinib + venetoclax (**A+V**), was evaluated in the AMPLIFY trial (NCT03836261)[8], which is a randomized, global, multi-centre, open-label Phase III study comparing **A+V** to investigator's choice of chemoimmunotherapy (FCR or BR) in previously untreated CLL patients.
 - The **A+V** regimen consistently demonstrated a lower within-trial incidence of grade ≥3 adverse events (AEs) than FCR or BR.[8]

Objective

This study aimed to investigate and summarize published safety profiles of current 1L CLL treatments relevant to the Canadian setting

- The use of chemoimmunotherapy has become antiquated in Canada, with near-zero utilization, while targeted fixed-duration regimens have gained preference for their ability to reduce treatment time, minimize long-term toxicity, and improve patient compliance compared to TTP regimens[7,9]; therefore, this study focuses on targeted fixed-duration therapies.

Methods

- The literature review was conducted following standard methodologies adapted from the Cochrane Handbook for Systematic Reviews of Interventions.[10]
- Studies were identified in Embase (Ovid) from 2024 to May 5, 2025, using the PICO criteria (**Table 1**) to supplement a prior global systematic literature review (SLR); screening was performed by a methodologist with support from Evidinno's proprietary AI-enabled selection tool, and any discrepancies were resolved by consensus with a second reviewer. All eligible studies were included for data extraction.
- Additionally, manual searches of publicly available European Public Assessment Reports (EPARs) for treatment combinations of interest were conducted via the EMA website (https://www.ema.europa.eu/en/medicines).

Table 1: Simplified PICOs eligibility criteria

Parameter	Description
Population	Adults with CLL receiving 1L treatment
Intervention	A+V, I+V, V+O, ibrutinib monotherapy, acalabrutinib monotherapy, and zanubrutinib monotherapy
Comparator	Any treatment
Outcomes	TEAEs, TRAEs, AE-related treatment discontinuations, specific AEs of interest, and mortality
Study Design	Clinical trials, observational studies including RWE using real-world data, and systematic reviews and meta-analyses for library only
Language	English
Publication Date Limit	• 2024 - May 5, 2025 (clinical trials) • 2013 - May 5, 2025 (RWE)

Abbreviations: 1L = first-line; A+V = acalabrutinib + venetoclax; AEs = adverse events; CLL = chronic lymphocytic leukemia; I+V = ibrutinib + venetoclax; RWE = real-world evidence; TEAEs = treatment-emergent adverse events; TRAEs = treatment-related adverse events; V+O = venetoclax + obinutuzumab.

Results

- In total, 2,090 records were identified from the Embase database, of which 2,045 were screened at title/abstract screening stage following removal of duplicate records (**Figure 1**).
- After full-text screening of 146 publications, 10 publications representing 11 unique studies (one publication reported two separate clinical trials) were included:
 - 7 clinical trials
 - 4 observational studies
 - Additionally, two clinical trials were sourced from the global SLR

Figure 1: PRISMA flow diagram

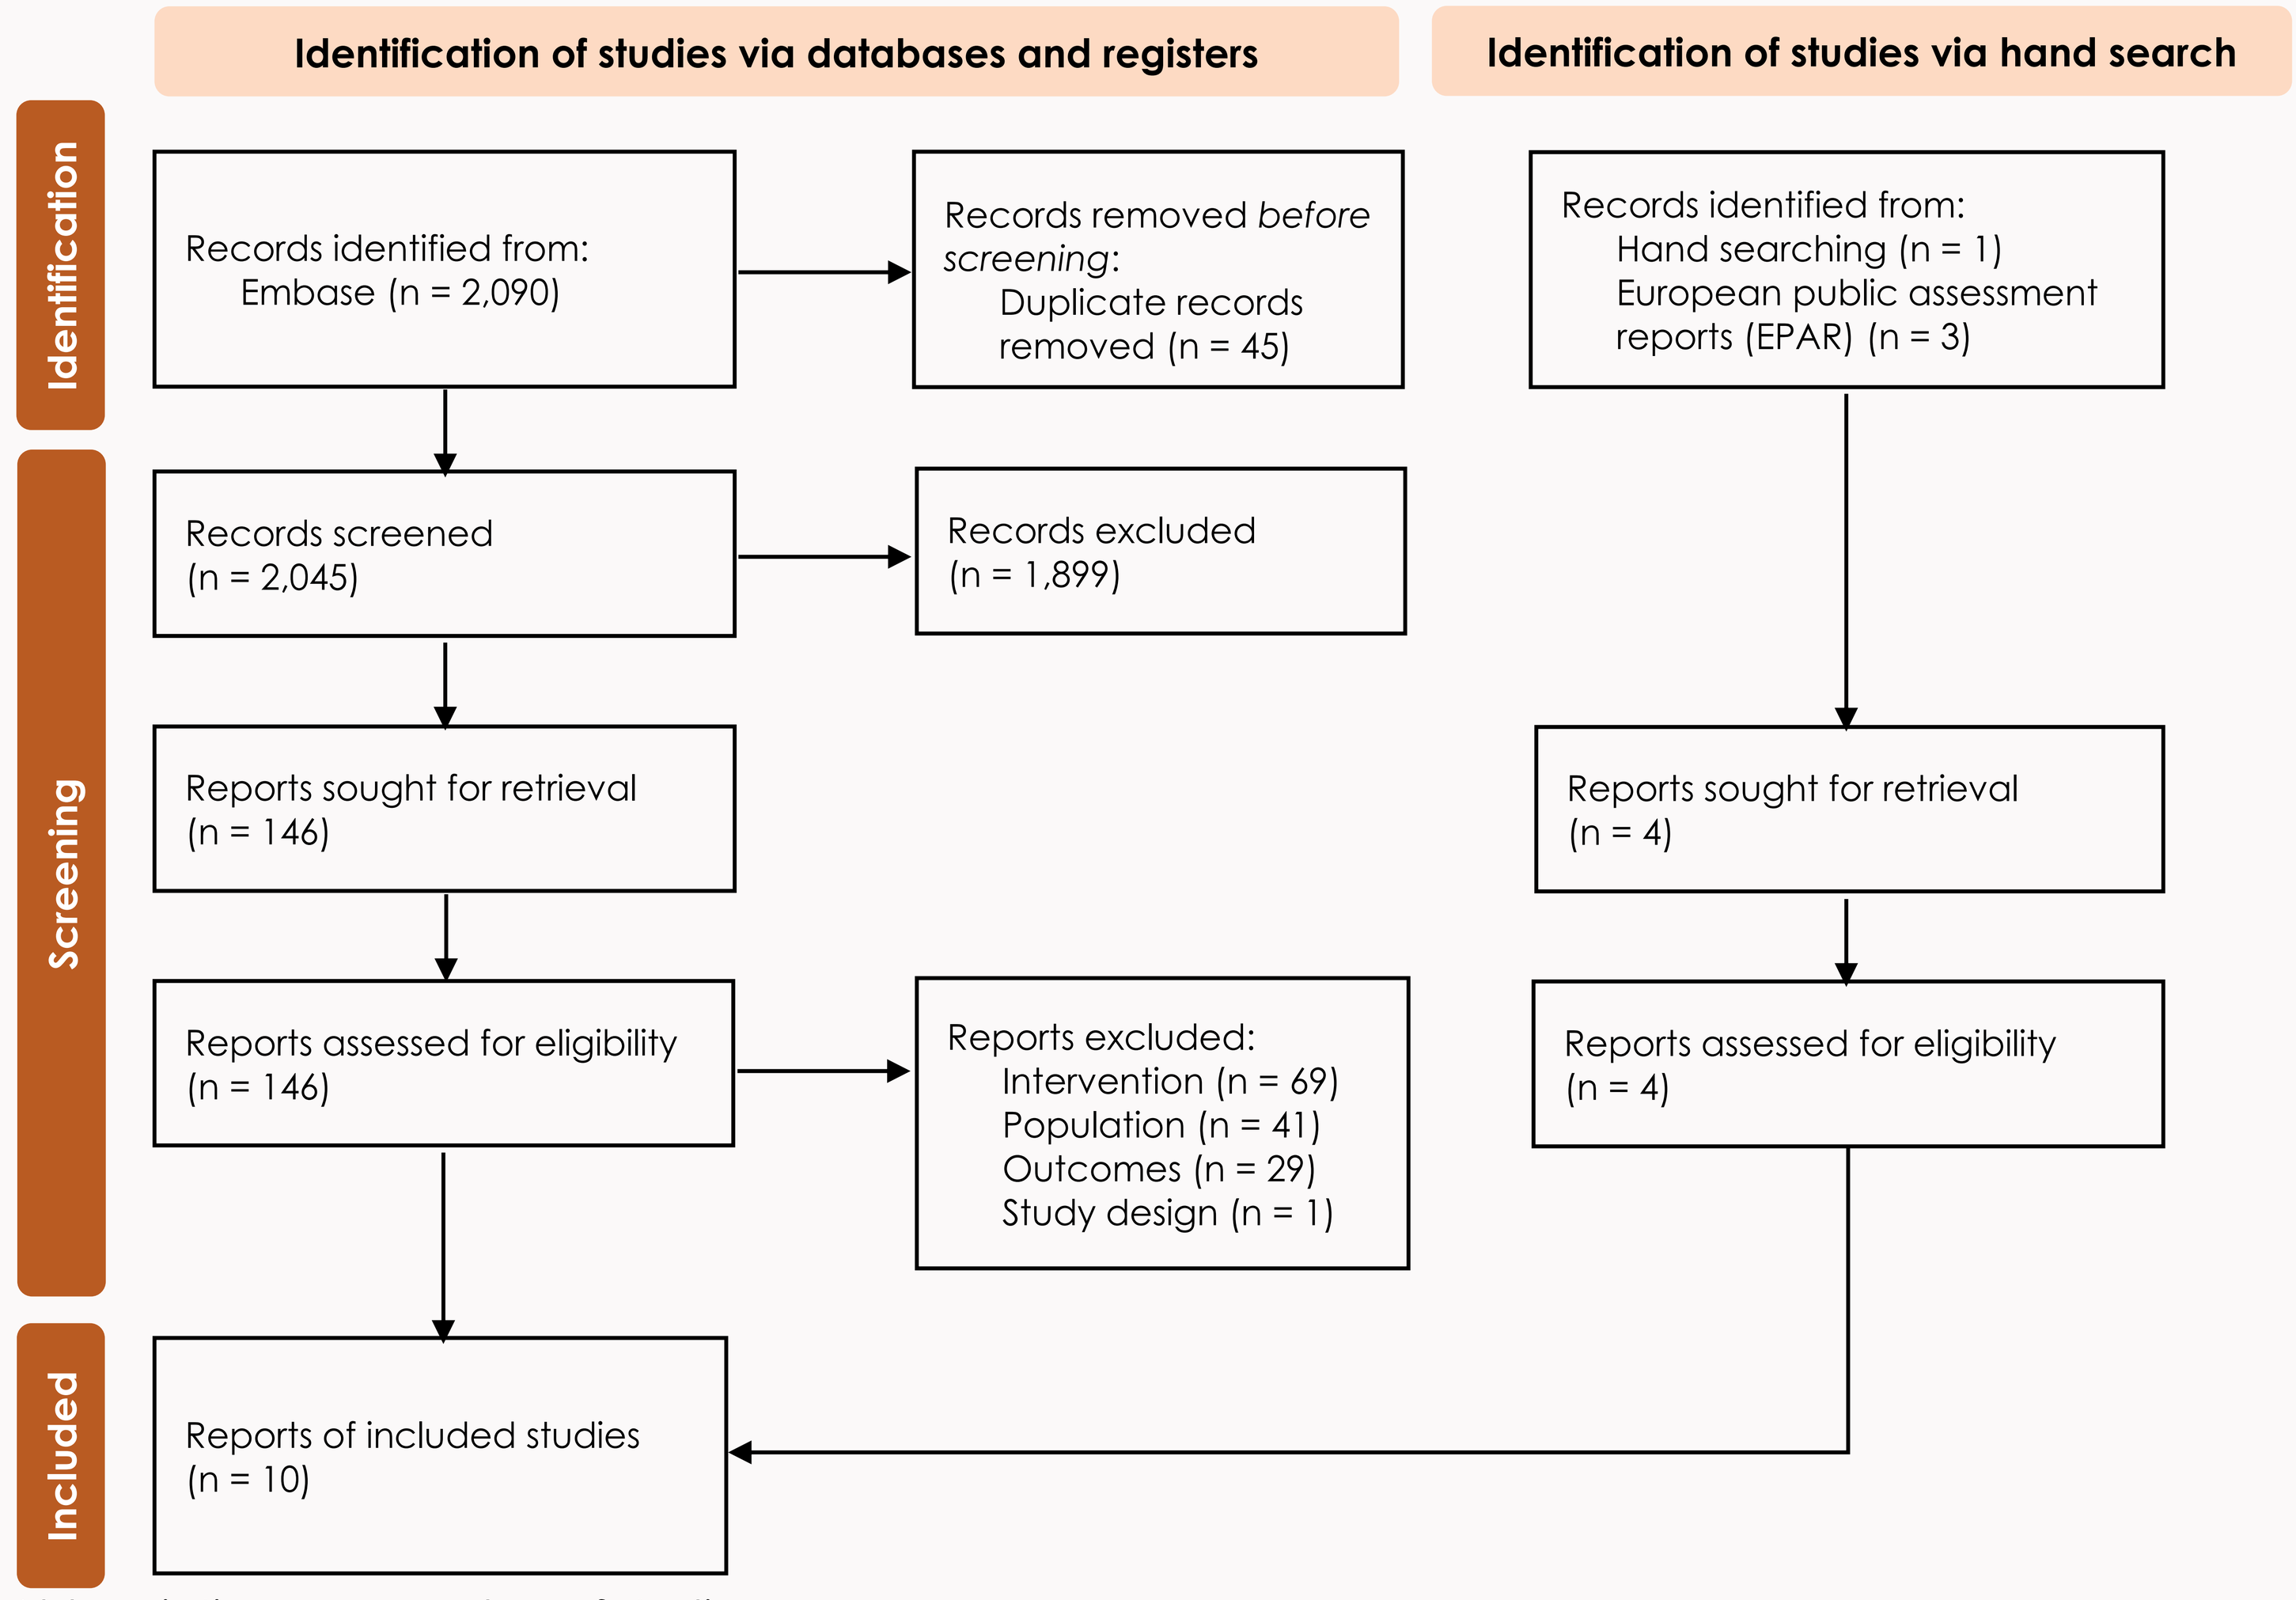
Identification of studies via databases and registers
Identification of studies via hand search
Identification
Screening
Included
Records identified from: Embase (n = 2,090)
Records removed before screening: Duplicate records removed (n = 45)
Records identified from: Hand searching (n = 1) European public assessment reports (EPAR) (n = 3)
Records screened (n = 2,045)
Records excluded (n = 1,899)
Reports sought for retrieval (n = 146)
Reports sought for retrieval (n = 4)
Reports assessed for eligibility (n = 146)
Reports excluded: Intervention (n = 69) Population (n = 41) Outcomes (n = 29) Study design (n = 1)
Reports assessed for eligibility (n = 4)
Reports of included studies (n = 10)

Abbreviations: n = number of studies.

- In the AMPLIFY trial, **A+V** demonstrated a manageable safety profile with a lower rate of grade ≥3 treatment-emergent AEs (TEAEs) at **53.6%** and serious TEAEs at **24.7%**[8], compared with in-trial rates for **I+V** (**64.7-75.5%** grade ≥3 TEAEs; **21.7%-46.2%** serious TEAEs)[11] and **V+O** (**78.8%-80.3%** grade ≥3 TEAEs; **44.7%-49.1%** serious TEAEs).[12,13]
- The **A+V** regimen reported the lowest within-trial rate of any treatment-related AEs (TRAEs) at **79.0%** and serious TRAEs at **9.3%** based on the AMPLIFY trial[8] compared with **I+V** (**84%-95%** any TRAEs; **12.1%-24.5%** serious TRAEs)[11] and **V+O** (**89.6%** any TRAEs; **26.4%** serious TRAEs).[12]
- The **A+V** regimen was associated with improved tolerability, reporting a lower within-trial rate of treatment discontinuation due to TEAEs (AMPLIFY: **7.9%**[8]) than rates observed for **I+V** (GLOW: **19.8%**[11]) and **V+O** (CLL14: **16.0%**[12]).
- The **A+V** regimen reported low within-trial rates of cardiac events, including any-grade cardiac AEs (**9.3%**) and grade ≥3 cardiac AEs (**1.7%**)[8], while a higher incidence was reported for **I+V** (**21.7%-24.5%** any-grade cardiac AEs; **3.4%-12.3%** grade ≥3 cardiac AEs)[11] and **ibrutinib monotherapy** (**14.8%** any-grade cardiac AEs; **5.9%** grade ≥3 cardiac AEs).[14]
- Atrial fibrillation was infrequently reported for the **A+V** regimen (**0.7%** any grade; **0.3%** grade ≥3)[8], representing a potential safety advantage over ibrutinib-containing regimens, whereas higher within-trial incidences were observed with **I+V** (**5.9%-14.2%** any grade; **1.5-6.6%** grade ≥3)[11], and **ibrutinib monotherapy** (**7.4%** any grade; **2.2%** grade ≥3).[14]
- In the AMPLIFY trial[8], **A+V** demonstrated low grade ≥3 rates of anemia, neutropenia, hypertension, infections, and tumor lysis syndrome reporting within-trial rates that were similar to or lower than those observed with **I+V**[11] and notably lower than with **V+O**[12,13] (**Table 2**).
- In the AMPLIFY trial, **A+V** (an all-oral regimen) had **zero** infusion-related reactions, whereas **obinutuzumab**-containing combinations reported incidence rates of any-grade events up to **51.3%**.[13]

Table 2: Specific AEs of interest

Grade ≥3 Adverse events, n (%)	AMPLIFY – Safety population[8] Median follow-up: 40.88 months	CAPTIVATE – Safety population[11] Median follow-up: 27.9 months	GLOW – Safety population[11] Median follow-up: 27.7 months	CLL14 – Safety population[12] Median follow-up: 28.1 months	CLL13 – Safety population[13] Median follow-up: 38.8 months
	A+V (n = 291)	I+V (n = 323)	I+V (n = 106)	V+O (n = 212)	V+O (n = 228)
Anemia	11 (3.8%)	6 (1.9%)	3 (2.8%)	17 (8.0%)[15]	11 (4.8%)
Neutropenia	94 (32.3%)	110 (34.1%)	30 (28.3%)	112 (52.8%)	103 (45.2%)
Hemorrhage	3 (1.0%)	3 (0.9%)	4 (3.8%)	-	-
Hypertension	8 (2.7%)	16 (7.1%)	9 (8.5%)	6 (2.8%)	4 (1.8%)
Infection	36 (12.4%)	27 (8.4%)	16 (15.1%)	41 (19.3%)	30 (13.2%)
Infusion-related reaction	0	0	0	19 (9.0%)[15]	26 (11.4%)
Tumor lysis syndrome	1 (0.3%)	1 (0.3%)	0	-	19 (8.3%)

Abbreviations: A+V = acalabrutinib + venetoclax; I+V = ibrutinib + venetoclax; n = number of patients; V+O = venetoclax + obinutuzumab.
NOTE: These data are derived from different studies with varying designs, patient populations, AE definitions, and follow-up durations. Direct comparisons should be interpreted with caution due to methodological differences between studies.

- In the AMPLIFY trial, **A+V** had an all-cause mortality rate of **6.2%**, with more than half of the deaths related to COVID-19 rather than the treatment itself[8]; deaths due to TEAEs were **3.4%**[8] (mostly COVID-19-related). This was a lower reported within-trial incidence than **I+V** in GLOW (**6.6%**)[11] and higher than **V+O** in CLL14 (**1.9%**[12]), which was not conducted during the COVID-19 pandemic.
- Four RWE studies on 1L CLL treatment focused on **ibrutinib monotherapy**, with none evaluating combination therapies

Conclusions

- Although comparisons are limited by cross-trial heterogeneity, the **A+V** regimen consistently demonstrated lower within-trial rates of grade ≥3 AEs, serious TEAEs, and key AEs of clinical interest - including cardiac AEs, infusion-related reactions, and tumor lysis syndrome compared with other fixed duration combination therapies (**I+V** and **V+O**) available in Canada.
- Taken together, these findings support **A+V** as a potential treatment option for 1L CLL treatment, given its manageable safety profile.
- Future head-to-head trials and real-world studies will be important to validate these findings and further strengthen **A+V**'s positioning in the evolving Canadian CLL treatment landscape.

References

1. Kittai AS, Huang Y, Miller S, et al. Outcomes of patients with Richter transformation who received no prior chemoimmunotherapy for their CLL. Blood Cancer Journal 2025;15(1):23. DOI: 10.1038/s41408-025-01236-6.
2. Statistics Canada. Table: 13-10-0111-01: Number and rates of new cases of primary cancer, by cancer type, age group and sex. (https://www150.statcan.gc.ca/t1/tbl1/en/tv.action?pid=1310011101).
3. Canadian Cancer Society. Chronic lymphocytic leukemia statistics. (https://cancer.ca/en/cancer-information/cancer-types/chronic-lymphocytic-leukemia-cll/statistics).
4. Statistics Canada. Table 13-10-0142-01 Deaths, by cause, Chapter II: Neoplasms (C00 to D48). (https://www150.statcan.gc.ca/t1/tbl1/en/tv.action?pid=1310014201).
5. Lachaine J, Guinan K, Aw A, Banerji V, Fleury I, Owen C. Impact of Fixed-Duration Oral Targeted Therapies on the Economic Burden of Chronic Lymphocytic Leukemia in Canada. Current oncology (Toronto, Ont) 2023;30(5):4483-4498. (In eng). DOI: 10.3390/curroncol30050339.
6. Owen C, Banerji V, Johnson N, et al. Canadian evidence-based guideline for frontline treatment of chronic lymphocytic leukemia: 2022 update. Leukemia research 2023;125:107016. (In eng). DOI: 10.1016/j.leukres.2023.107016.
7. Cancer Care Alberta. Chronic lymphocytic leukemia – Clinical Practice Guideline LYHE-007, Version 11. Alberta Health Services. (https://www.albertahealthservices.ca/assets/info/hp/cancer/if-hp-cancer-guide-lyhe007-cll.pdf).
8. Brown JR, Seymour JF, Jurczak W, et al. Fixed-Duration Acalabrutinib Combinations in Untreated Chronic Lymphocytic Leukemia. The New England journal of medicine 2025;392(8):748-762. (In eng). DOI: 10.1056/NEJMoa2409804.
9. Pullen LC. Fixed-Duration Regimen Safe, Effective for CLL. ASH Clinical News. (https://ashpublications.org/ashclinicalnews/news/8053/Fixed-Duration-Regimen-Safe-Effective-for-CLL).
10. Higgins JPT, Thomas J, Chandler J, et al. Cochrane Handbook for Systematic Reviews of Interventions version 6.5 (updated August 2024). 2024; www.cochrane.org/handbook.
11. European Medicines Agency. Committee for Medicinal Products for Human Use. Assessment Report. Imbruvica. (https://www.ema.europa.eu/en/documents/variation-report/imbruvica-h-c-003791-ii-0070-epar-assessment-report-variation_en.pdf).
12. European Medicines Agency. Committee for Medicinal Products for Human Use. CHMP group of variations including an extension of indication assessment report. Venclyxto. (https://www.ema.europa.eu/en/documents/variation-report/venclyxto-h-c-4106-ii-0023-g-epar-assessment-report-variation_en.pdf).
13. Eichhorst B, Niemann CU, Kater AP, et al. First-Line Venetoclax Combinations in Chronic Lymphocytic Leukemia. New England Journal of Medicine 2023;388(19):1739-1754. (Article) (In English). DOI: 10.1056/NEJMoa2213093.
14. European Medicines Agency. Committee for Medicinal Products for Human Use. Assessment Report. Imbruvica. (https://www.ema.europa.eu/en/documents/variation-report/imbruvica-h-c-3791-ii-00016-epar-assessment-report-variation_en.pdf).
15. Fischer K, Al-Sawaf O, Bahlo J, et al. Venetoclax and obinutuzumab in patients with CLL and coexisting conditions. New England Journal of Medicine 2019;380(23):2225-2236. (Article) (In English). DOI: 10.1056/NEJMoa1815281.

Acknowledgements

- This study was conducted by Evidinno Outcomes Research Inc., funded by AstraZeneca Canada
- This poster was created by AstraZeneca Canada in collaborations with Evidinno Outcomes Research Inc.